DEPLOYING THE GENEDATA PROFILER PLATFORM FOR VALUE CREATION IN DEBIOPHARM'S DRUG DEVELOPMENT PROCESS

Examples of Debiopharm data integration and meta-analysis

Simone Ragusa, Jeannette Fuchs, Hélène Cannelle, Franck Brichory, Carolina Haefliger

SUMMARY

Debiopharm is a Swiss biopharmaceutical company developing novel therapies to target high unmet medical needs in oncology and bacterial infectious diseases. Digitalization is fundamental for Debiopharm corporate. Debiopharm International, the company branch focused on drug development, has recently adopted Genedata Profiler (PRO) as a key platform for leveraging translational data value.

We are integrating PRO in the Translational Medicine Department (TM), adapting data structure with the long-term objective of using the platform as our data analysis tool for our research and development. Here we present examples of how PRO is being used for value creation on our data: the speed and precision in data validation, harmonization of data analysis, increased depth of analysis, and the possibility to create preclinical meta-analyses.

BACKGROUND

Debiopharm relies on both insourced and outsourced studies for research and development. Our scientists co-ordinate the data generation with specialized vendors, which ultimately imprint results and reports in their respective formats and with their own interpretations. Although this process complies with compound requirements, scientist can collect only partial and not-standardized data, limiting their capitalization. Debiopharm wishes to collect, standardize, structure, store, manage, and analyse data internally to increase data consumption and value. PRO is set to be a fundamental player in Debiopharm digitalization strategy, and it will complement our current AI and digital approaches, for state-of-the-art drug development

METHODS

To ensure the data from various sources aligns with Debiopharm's structure and template, we have initiated within TM the development of customized integration workflows (WFs). This will allow us in the long term to establish a validated PRO production environment. Currently, Debiopharm integration WFs fall into two main fields: data validation and virtual meta-analysis (multiple studies and data types).

In parallel, Debiopharm is developing two initiatives that will also further support PRO deployment:

- A Central Data Repository (CDR), for centralized data storage, in direct connection to PRO, ensuring the integrity of data to be analysed.
- A Metadata Repository (MDR), to enhance data governance by automated quality control accordingly to Debiopharm metadata, to also ensure standardization of entire programs in PRO.

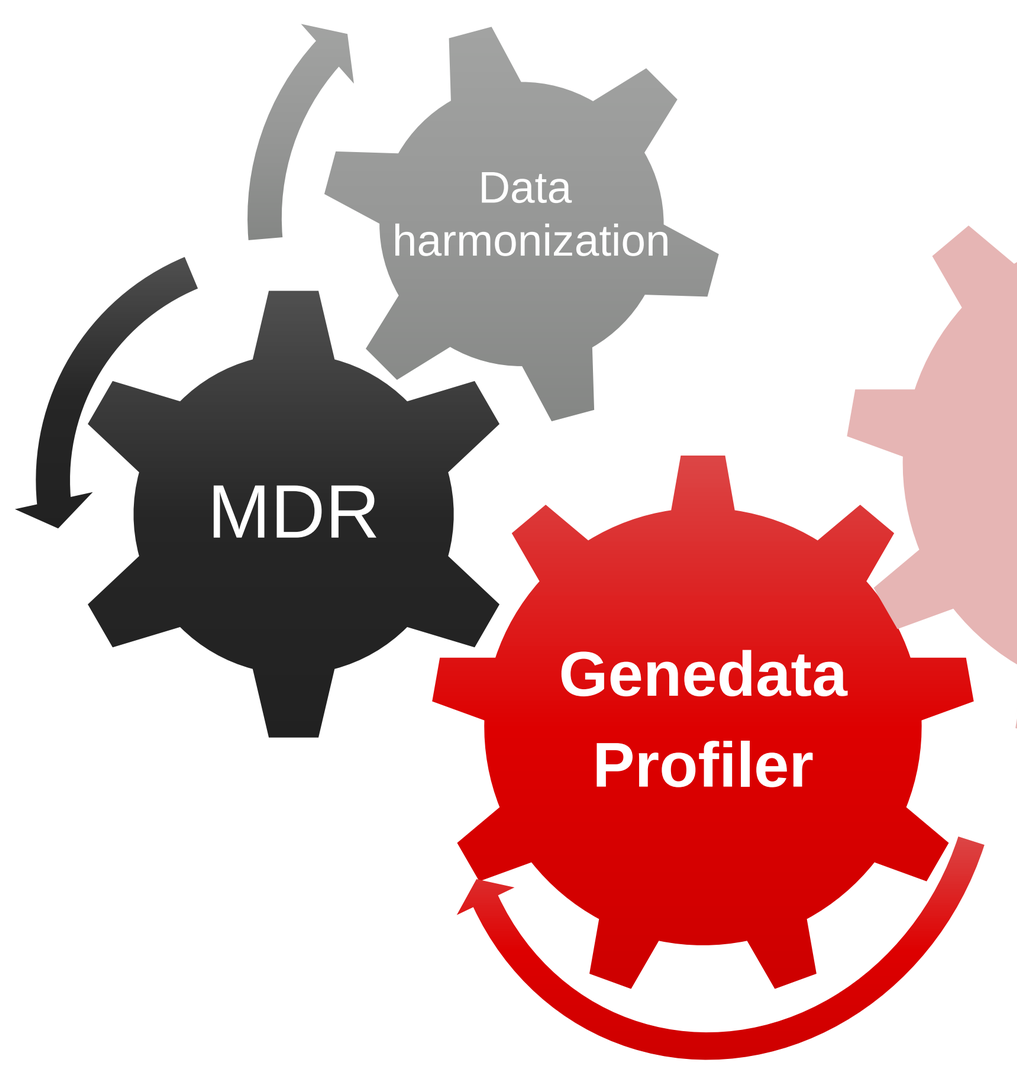
Data harmonization
MDR
CDR
Genedata Profiler

RESULTS

Standardization and consumption of data, into Debiopharm' pipeline

Debiopharm is currently focusing on data standardization. Together with creation of assays integration WFs, we prioritized two tasks fundamental for comparability of differently sourced data: the harmonization of Raw Sequencing data (Figure 1A), and the calculation of standardized data (e.g. IC50) from different studies and vendors (Figure 1B). Additionally, we have designed customized dashboards to facilitate the visualization of toxicology datasets standardized with CDISC SEND nomenclature (Figure 1C).

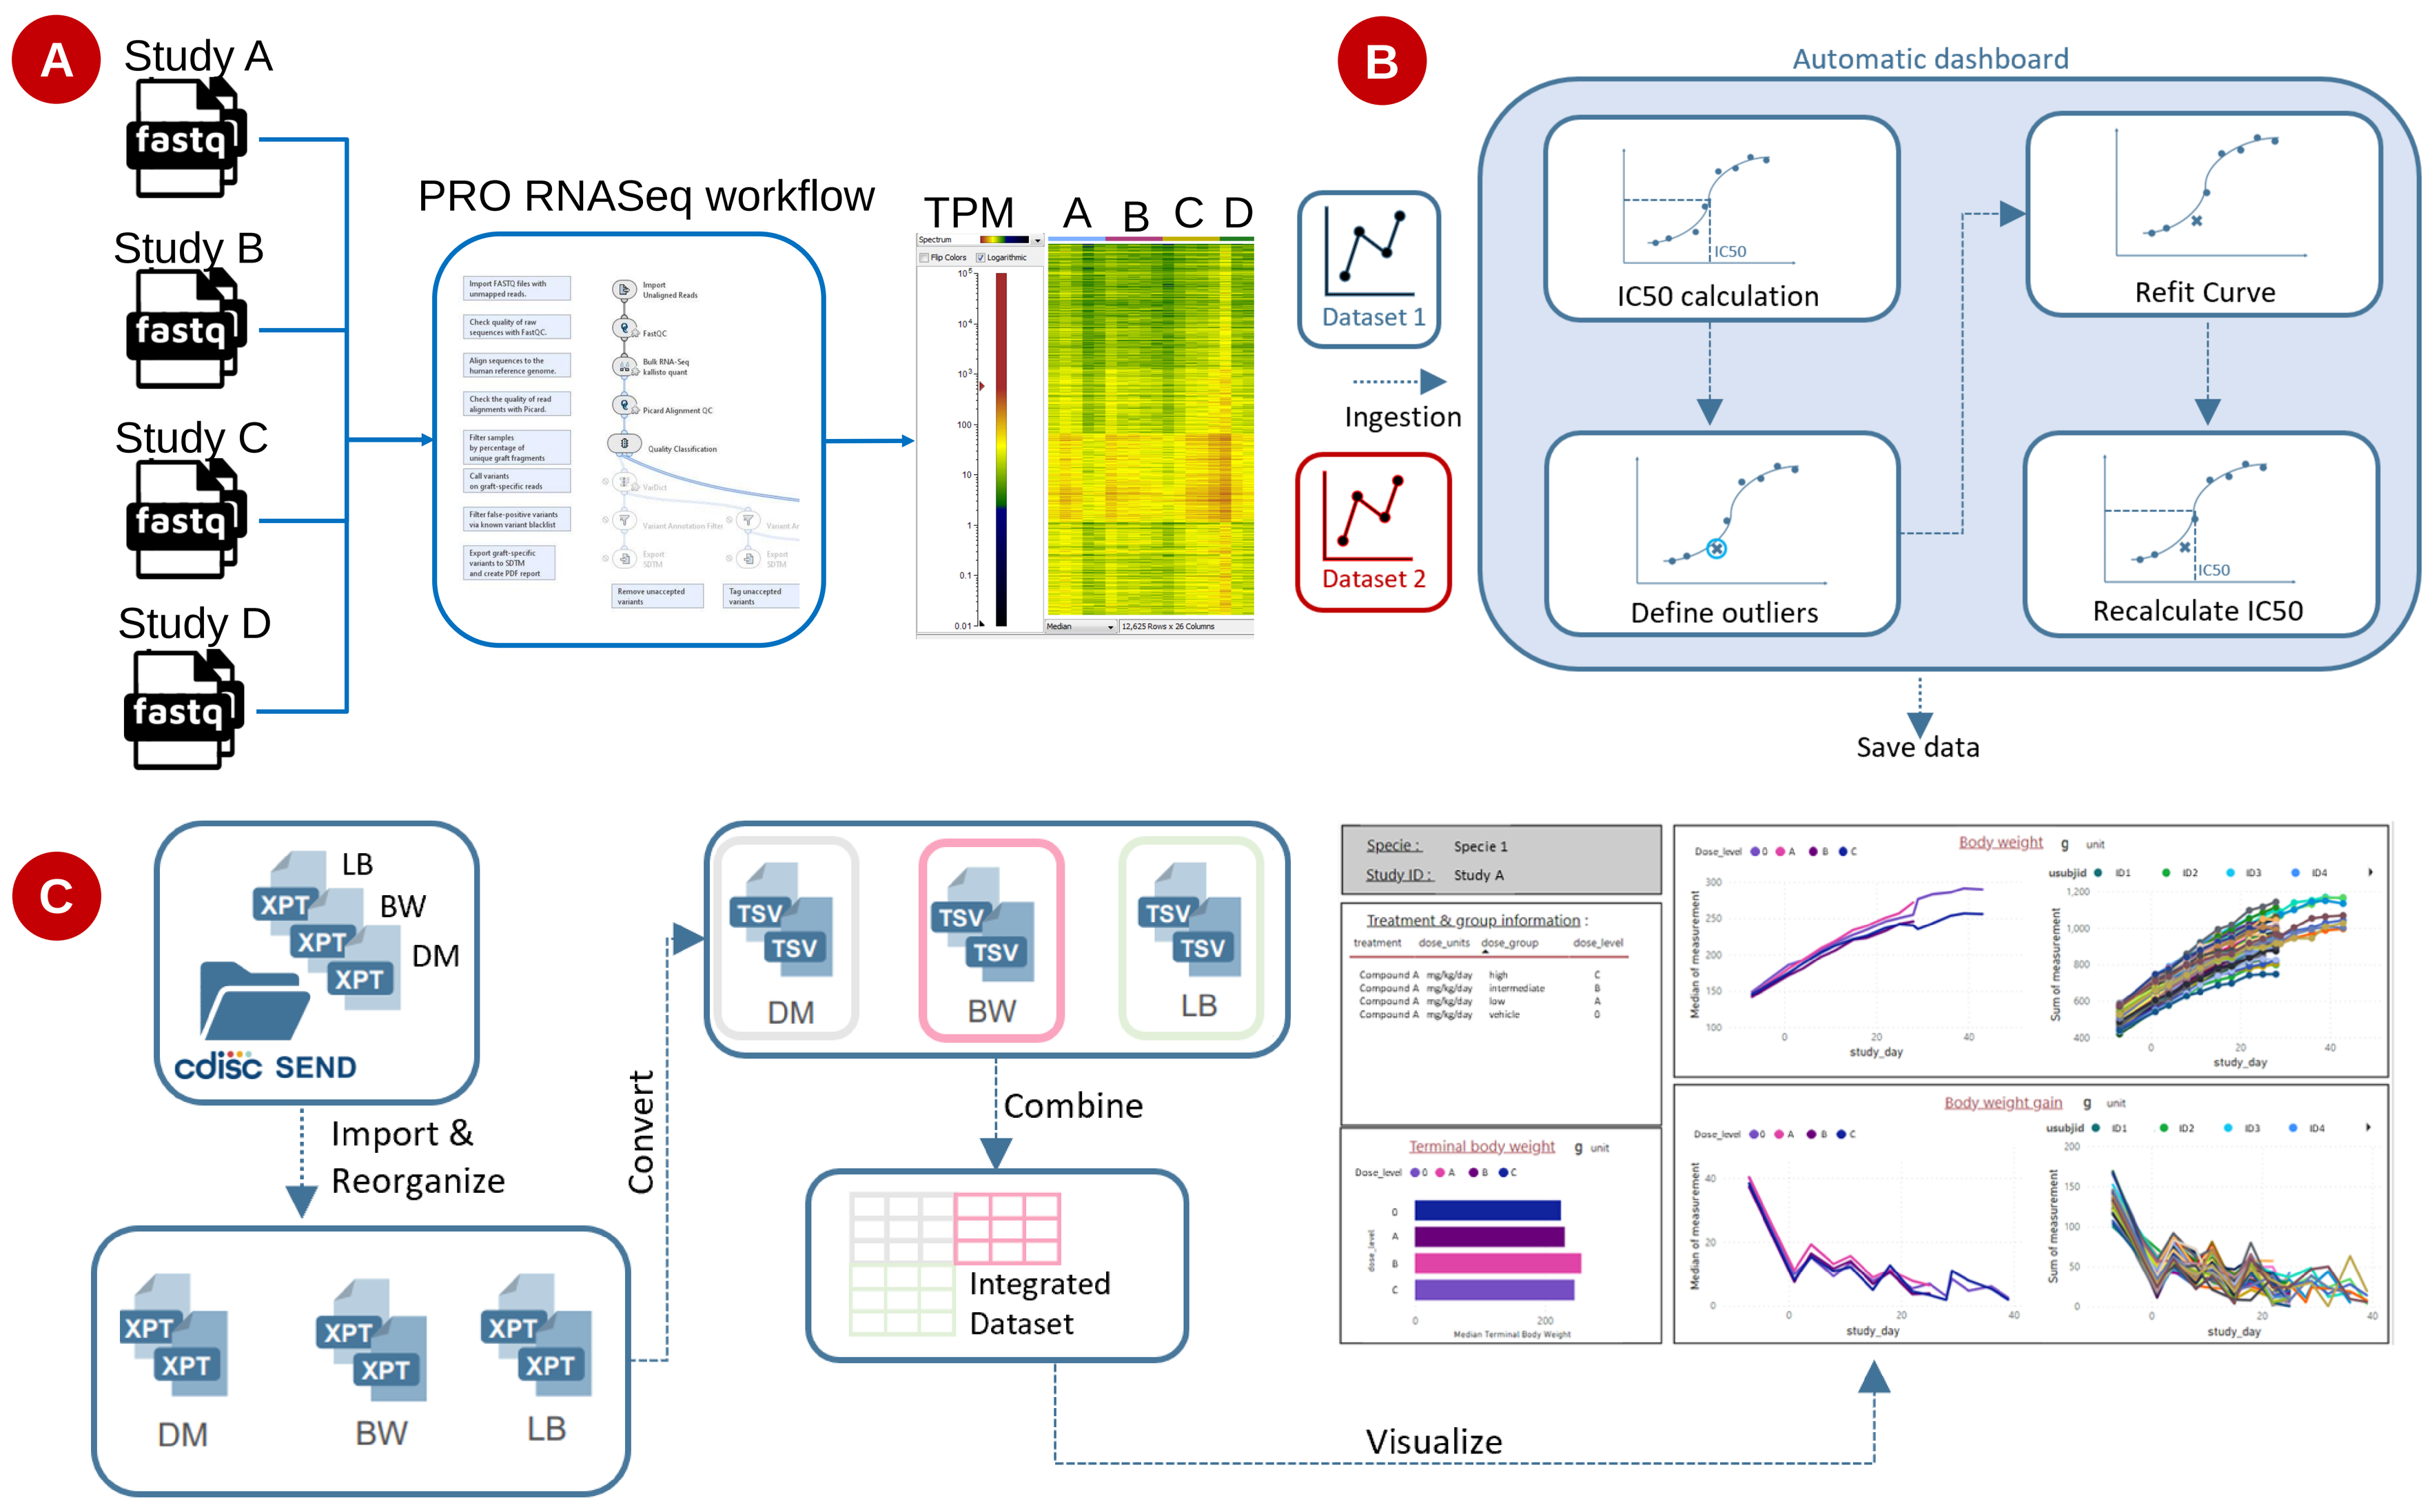
A
Study A
Study B
Study C
Study D
fastq
PRO RNASeq workflow
TPM
A B C D
B
Automatic dashboard
Dataset 1
Ingestion
Dataset 2
IC50 calculation
Refit Curve
Define outliers
Recalculate IC50
Save data
C
LB
BW
DM
cdisc SEND
Import &
Reorganize
Convert
Combine
Integrated
Dataset
Visualize
Body weight
Terminal body weight
Body weight gain

Figure 1. Processes deployed for ensuring Debiopharm data standardization in PRO **(A)** Example Scheme, RNA Seq ingestion WF, exploiting Kallisto quant pseudo-alignment process for host-contamination cleaning and normalization of bulk transcriptomic NGS data from cancer cell line derived xenograft models **(B)** Example Scheme IC50 calculation, tool under development with PRO team to support and harmonize screening IC50 calculations and flagging of data outliers **(C)** Automatic workflows for toxicology reports with CDISC SEND data format to import, convert. and combine all dataset in PRO. The output is visualized in a customized dashboard designed on Power BI tool, and automatically connected to PRO.

Genedata Profiler Data Validation, to improve data quality

PRO allows Debiopharm TM users to quickly QC large datasets, independently from the data type. Vendors calculations, methods, and metadata are co-ingested. Outliers, and eventual calculation or experimental errors are identified in PRO in-built statistical tool, Analyst (Figure 2A). Moreover, meta-analysis allows data validation, comparing results with public data or additional Debiopharm data (Figure 2B).

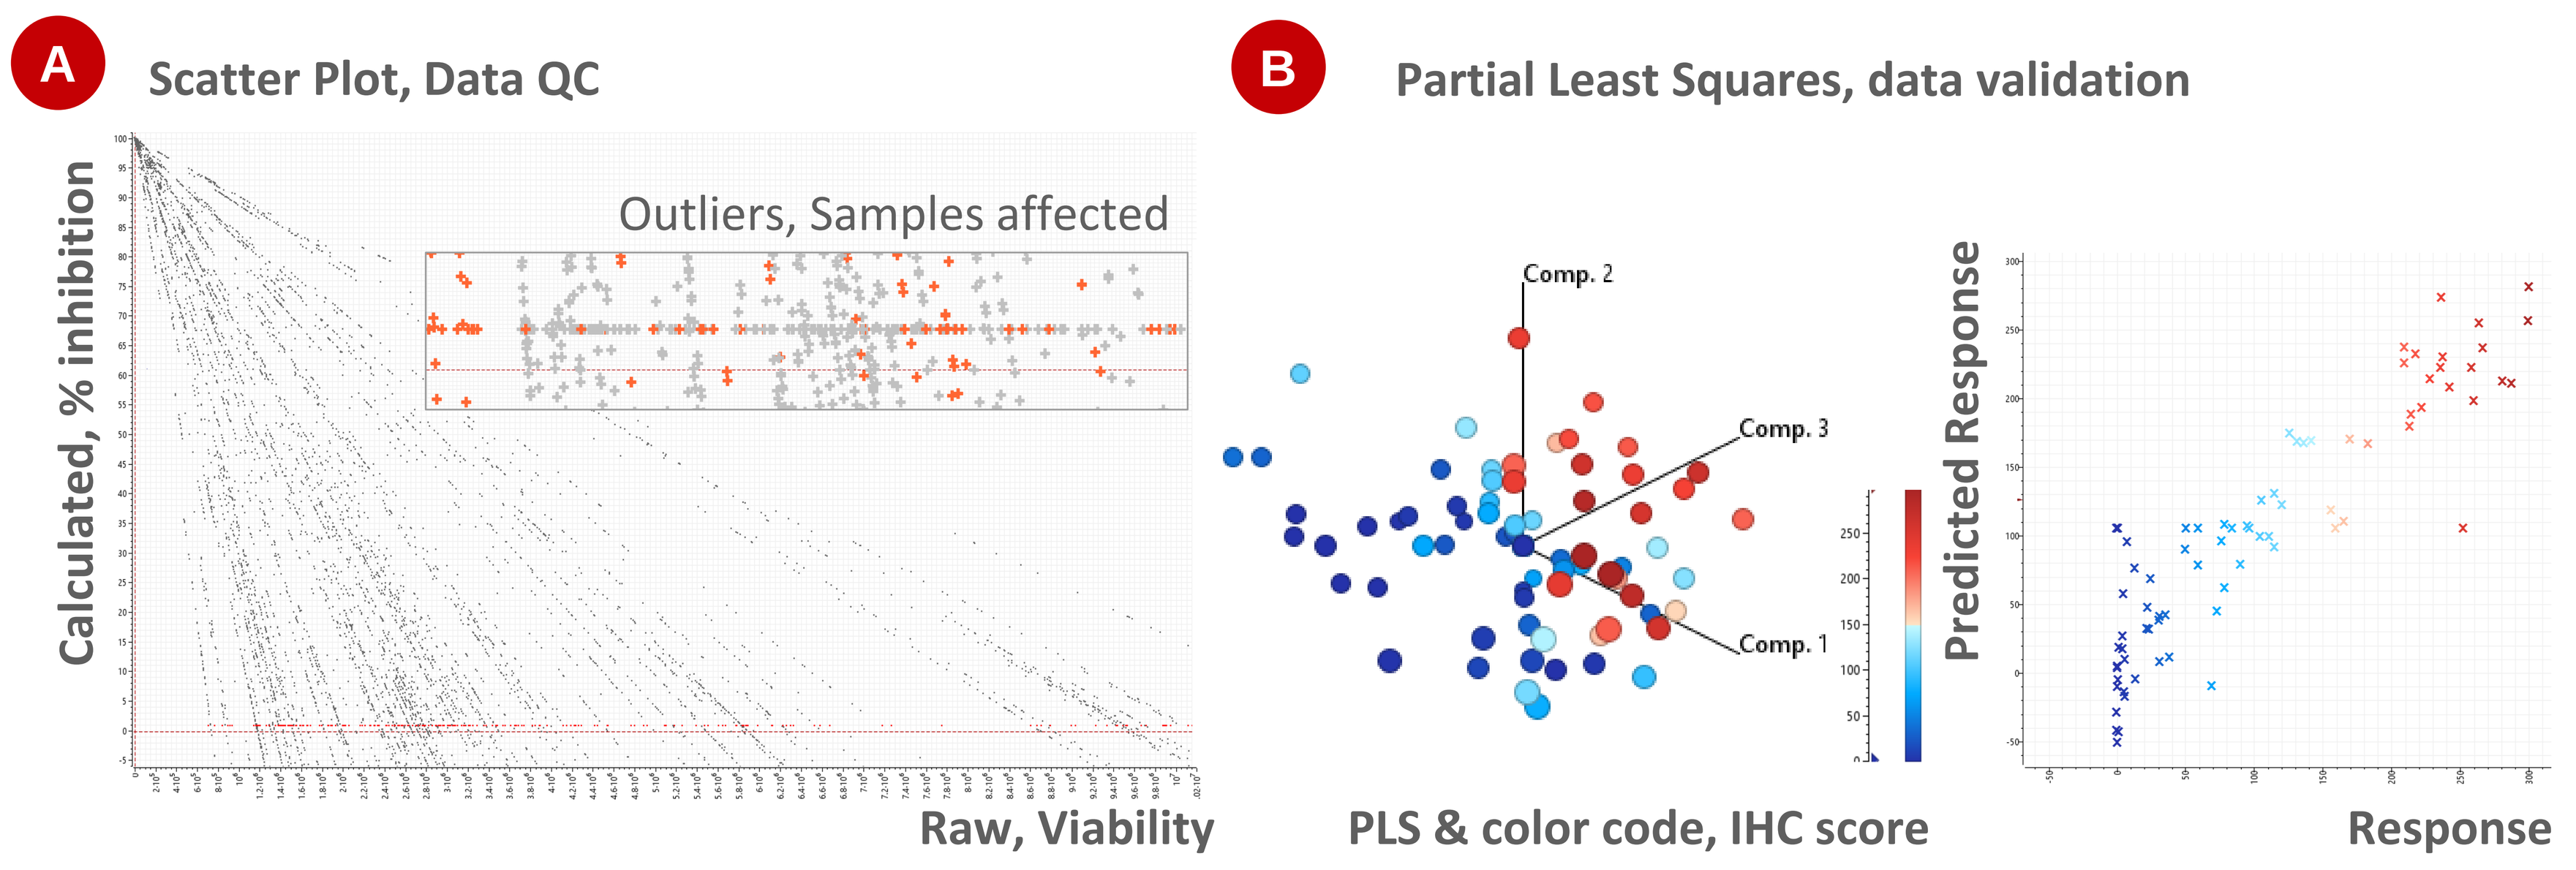
A
Scatter Plot, Data QC
Outliers, Samples affected
Calculated, % inhibition
Raw, Viability
B
Partial Least Squares, data validation
Comp. 2
Comp. 3
Comp. 1
PLS & color code, IHC score
Predicted Response
Response

Figure 2. Examples of Data Validation in PRO Analyst **(A)** Scatter Plot, data QC. Patient derived organoids *in vitro* viability screening upon treatment with Debiopharm therapeutic compound. Raw Data (x axis, bioluminescence detection), plotted against vendor calculations (% of growth inhibition over untreated control). Red dots in main plot indicate the control-normalization samples. In the zoomed plot, orange dots highlight samples with non-fitting normalization. **(B)** Partial Least Squares analysis of cancer cell lines transcriptome (RNASeq), calculated over the immunohistochemistry (IHC) score of a Debiopharm target molecule, on the correspondent *in vivo* cancer cell line derived xenograft models. On the left, PLS with IHC score represented by color code; on the right, Quality plot of IHC score based (x axis), PLS prediction (y axis).

RESULTS

Genedata Profiler co-analysis of screening data and basal sample transcriptomics, to better inform and prioritize conclusions

PRO allows to co-analyse deep sequencing data with study summary data. Example of a patient-derived organoid *in vitro* screening where viability treatment scores were co-analysed with the basal mRNA Sequencing from the samples. The screening included 2 indications, treated with one Debiopharm candidate drug. As expected, the global PCA analysis was dominated by tumor type (Figure 3A). Differently, Microarray (Figure 3B) or Limma GS (Figure 3C) analyses, on each individual indication, showed significant differential gene expression results valuable for biomarker discovery.

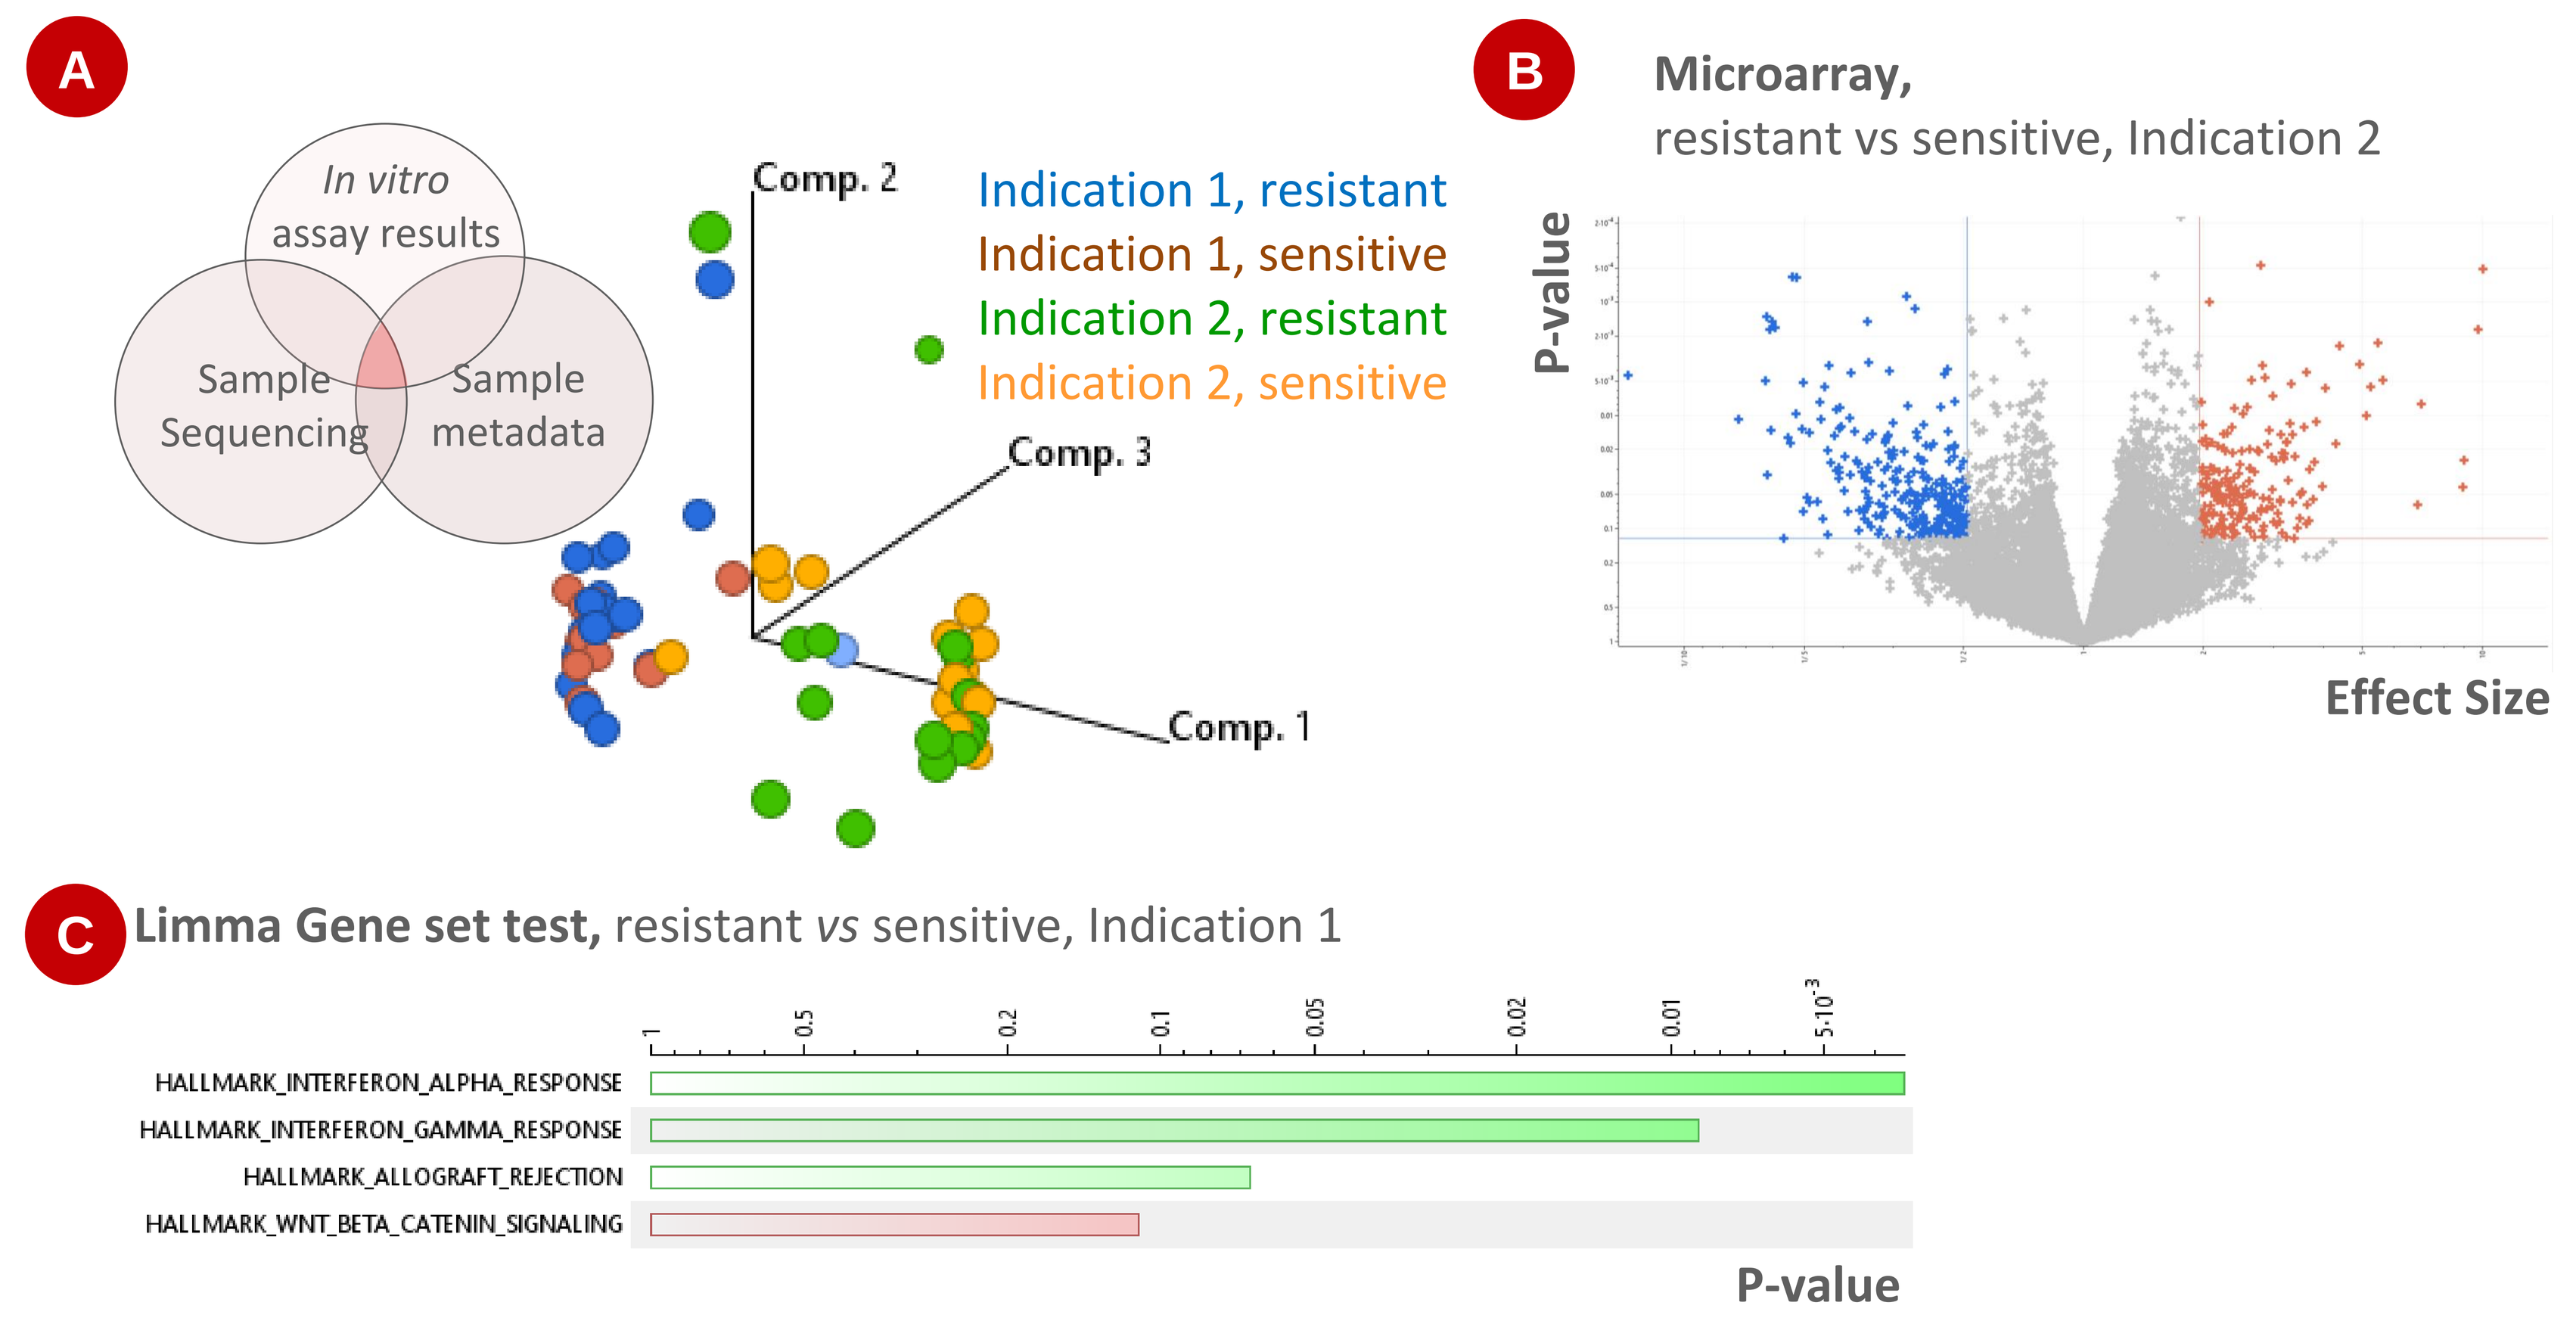
A
In vitro assay results
Sample Sequencing
Sample metadata
Comp. 2
Comp. 3
Comp. 1
Indication 1, resistant
Indication 1, sensitive
Indication 2, resistant
Indication 2, sensitive
B
Microarray,
resistant vs sensitive, Indication 2
P-value
Effect Size
C
Limma Gene set test, resistant vs sensitive, Indication 1
HALLMARK_INTERFERON_ALPHA_RESPONSE
HALLMARK_INTERFERON_GAMMA_RESPONSE
HALLMARK_ALLOGRAFT_REJECTION
HALLMARK_WNT_BETA_CATENIN_SIGNALING
P-value

Figure 3 Example of Debiopharm patient-derived organoids *in vitro* screening, co-analyzed with basal transcriptomics of samples **(A)** On the left, Study scheme. Efficacy screening data scores, sample mRNA sequencing and sample metadata were co-analyzed. On the right, PCA of all samples analyzed, from two different indications, both with encountered resistant and sensitive samples, to Debiopharm tested drug (color-code as indicated) **(B)** Volcano plot from Microarray test between resistant and sensitive samples from Indication 2 (blinded gene annotations) **(C)** Head results from Limma GS analysis on Cancer Hallmark Pathways between resistant and sensitive samples from Indication 1.

Genedata Profiler meta-analysis of standardized preclinical *in vivo* data, to improve translatability, and relevance of data package.

PRO allows meta-analysis of virtual-harmonized datasets across studies. Example of 3 independent *in vivo* studies which were co-analyzed, including efficacy, hematology, and PK data (Figure 4A). PRO comparative analysis confirmed global dose dependency (Figure 4B), but also supported compound prioritization, as shown by increased Neutrophils blood concentration as de-prioritization criterium, upon treatment with one of the three compounds (Figure 4C).

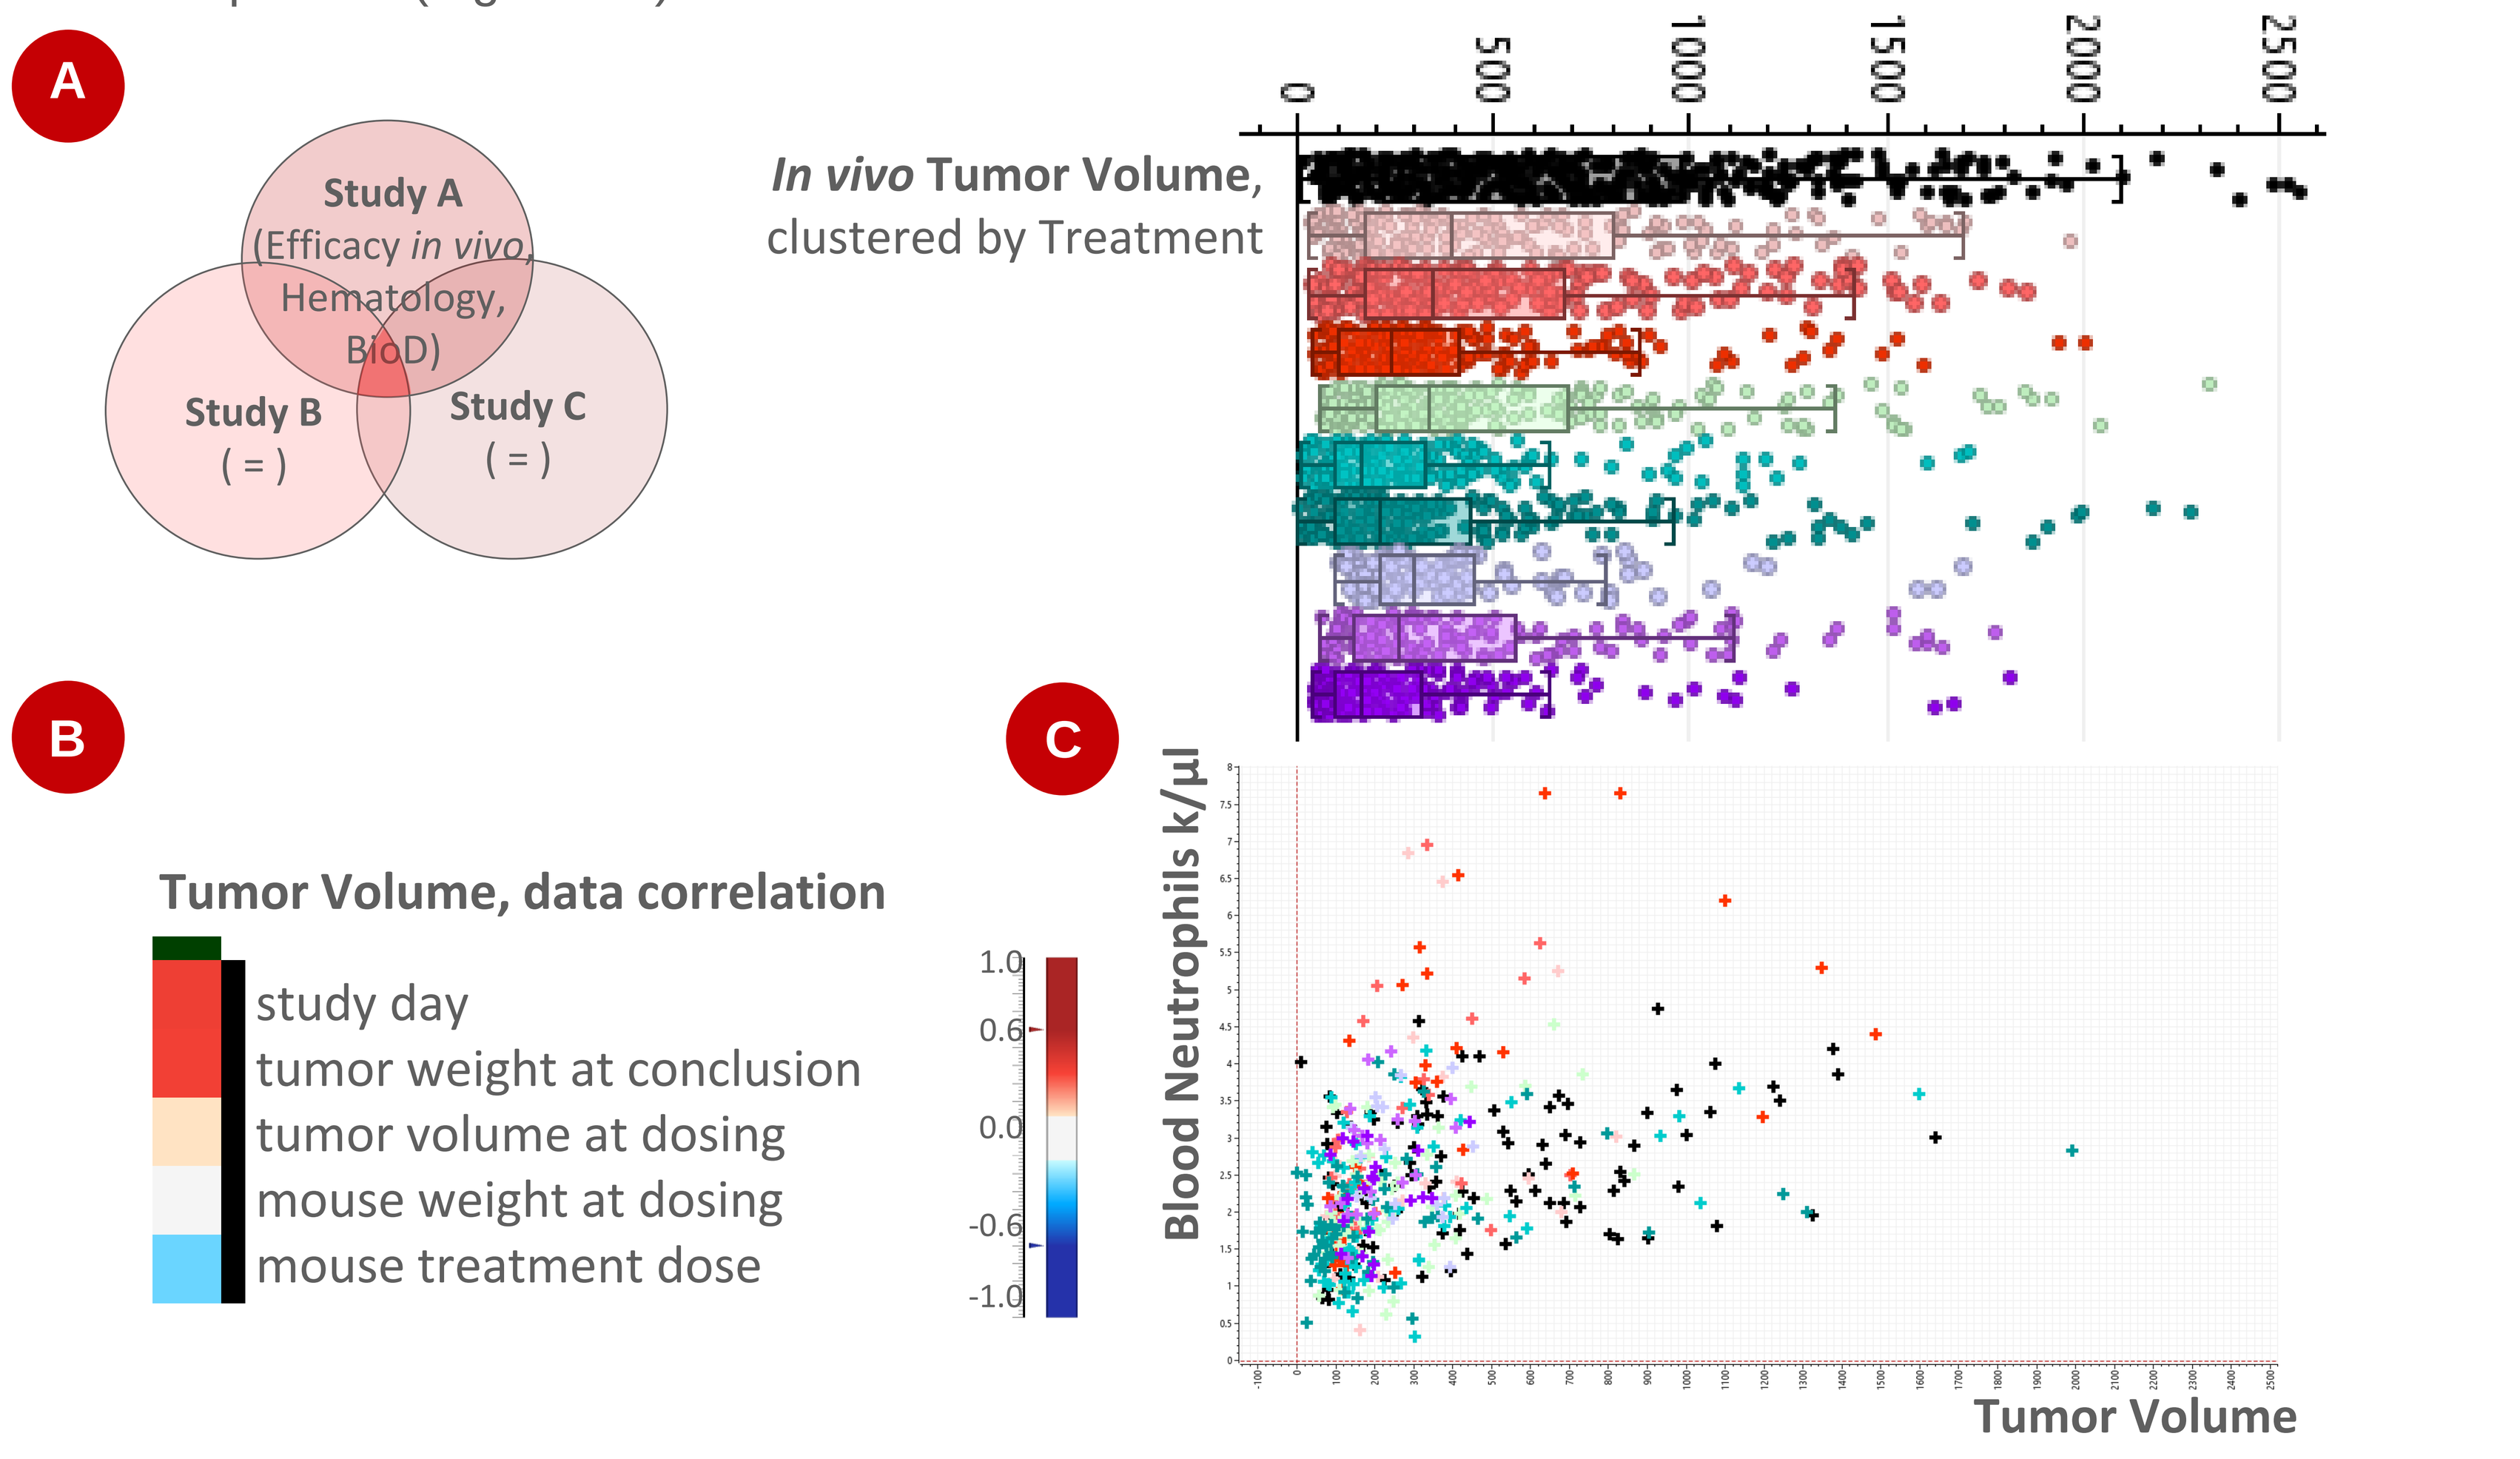
A
Study A
(Efficacy in vivo, Hematology, BioD)
Study B
(=)
Study C
(=)
In vivo Tumor Volume, clustered by Treatment
0
500
1000
1500
2000
2500
B
Tumor Volume, data correlation
study day
tumor weight at conclusion
tumor volume at dosing
mouse weight at dosing
mouse treatment dose
1.0
0.6
0.0
-0.6
-1.0
C
Blood Neutrophils k/µl
Tumor Volume

Figure 4. Example of Debiopharm *in vivo* CDX multi-study meta-analysis. **(A)** On the left, Study scheme. Efficacy Tumor Volume data, Hematology, and Biodistribution data, were co-analyzed from 3 *in vivo* studies. On the right, box plots of Tumor Volume measurements executed during *in vivo* monitoring, clustered by compound and dose treatment (dots color-code, black vehicle, red treated with compound 1, green compound 2, violet compound 3, color intensity represents dose levels) **(B)** Annotation correlation. Tumor volume measurements confirm positive correlation with study day and sample weight, while negative correlation with treatment dose levels **(C)** Annotations Scatter Plot: tumor volume (x axis), against Neutrophils blood concentration (y axis), for Hematology annotated measurements. Compound 1 flagged as increasing systemic Neutrophils.

ACKNOWLEDGEMENTS

We thank the PRO Supporting team, including Sebastien Ribrioux, Timo Wittenberger, Anahita Talebi Amiri, Hagen Klett, Pavan Kumar Nagaraj, and all others involved, for guiding us through the initial phases and the current Debiopharm implementation of PRO. We thank Debiopharm colleagues Noemie Luong, Norbert Wiedemann, Nicolas Quesnot, Vaibhav Kulkarni and Yannick Champion, that contributed to the integration and analysis of the shown Figures. Finally, we thank all other TM, DM and IT colleagues involved in PRO Platform Debiopharm development.

CONCLUSIONS

Debiopharm keeps digitalization at the core of its development.

In 2023 we demonstrated that PRO can significantly support TM scientists in data analysis, including multiple data co-analysis and in multi-study meta-analysis.

Debiopharm considers that the PRO platform has the potential to capitalize on the data relevance and translatability.

The company has the long-term objective of incorporating PRO as relevant platform for data analysis in Debiopharm.

CONTACT

Debiopharm International S.A.,
Lausanne, Switzerland.
www.debiopharm.com
Franck Brichory franck.brichory@debiopharm.com

DOWNLOAD

This poster is available via:
www.debiopharm.com/medias/publications