EHA 2024 POSTER P473

CD37 IS A RELEVANT TARGET FOR AML AND MDS TREATMENT WITH DEBIO 1562M ANTIBODY DRUG CONJUGATE (ADC)

Lisa Ivanschitz[1], Josée Hue-Perron[1], Selena Vigano[2], Sebastien Lofek[2], Noemie Luong[1], Léo Marx[2], Riccardo Colombo[1]

1: Debiopharm International SA, Chemin Messidor 5-7, CH-1006 Lausanne, Switzerland 2: Debiopharm Research & Manufacturing SA, Rue du Levant 146, CH-1920 Martigny, Switzerland

Debiopharm™ WE DEVELOP FOR PATIENTS

SUMMARY

CD37 relevance as an attractive therapeutic target for immune based therapies has been confirmed in clinical trials for B cell malignancies[1]. In acute myeloid leukemia (AML) and myelodysplastic syndrome (MDS), CD37 expression profile has been less investigated and is more controversial[2]. Here, we demonstrate that CD37 is broadly expressed in diverse AML and MDS models, including patient samples. Debio 1562M, a new CD37-targeting ADC, is efficiently internalized in these models, at a similar extent as in healthy or malignant B cells. With only one administration, Debio 1562M successfully triggers tumor regression of several cell-derived xenograft (CDX) models and strongly improves animal survival compared to venetoclax and azacitidine standard of care (SoC). In patient-derived xenograft (PDX) models, Debio 1562M significantly reduces tumor burden, similarly as venetoclax and azacitidine SoC treatment, or better in a SoC resistant model. In the absence of cross-reactive preclinical species, GLP toxicology study was performed in mice and demonstrated a safe profile related to known payload's toxicities.

BACKGROUND

CD37 is a trans-membrane protein, member of the tetraspanin superfamily, exclusively expressed on hematopoietic tissues. Primarily described on B cells, CD37 plays important roles in their development and modulating their immune function[3]. Increased expression of CD37 has been observed in various hematological cancers[4-7] and associated with poor patient outcome in AML[8-9].

Debio 1562M is a second-generation ADC directed against CD37, utilizing Debiopharm's proprietary Multilink™ cleavable linker technology. Eight molecules of a DM1 (mertansine) derivative, a highly potent anti-tubulin binder inducing mitotic catastrophe and cell death, are conjugated to naratuximab humanized antibody. Cathepsin-B-selective cleavable linker confers to Debio 1562M an excellent plasma stability allowing long exposure in mice and a safe toxicologic profile.

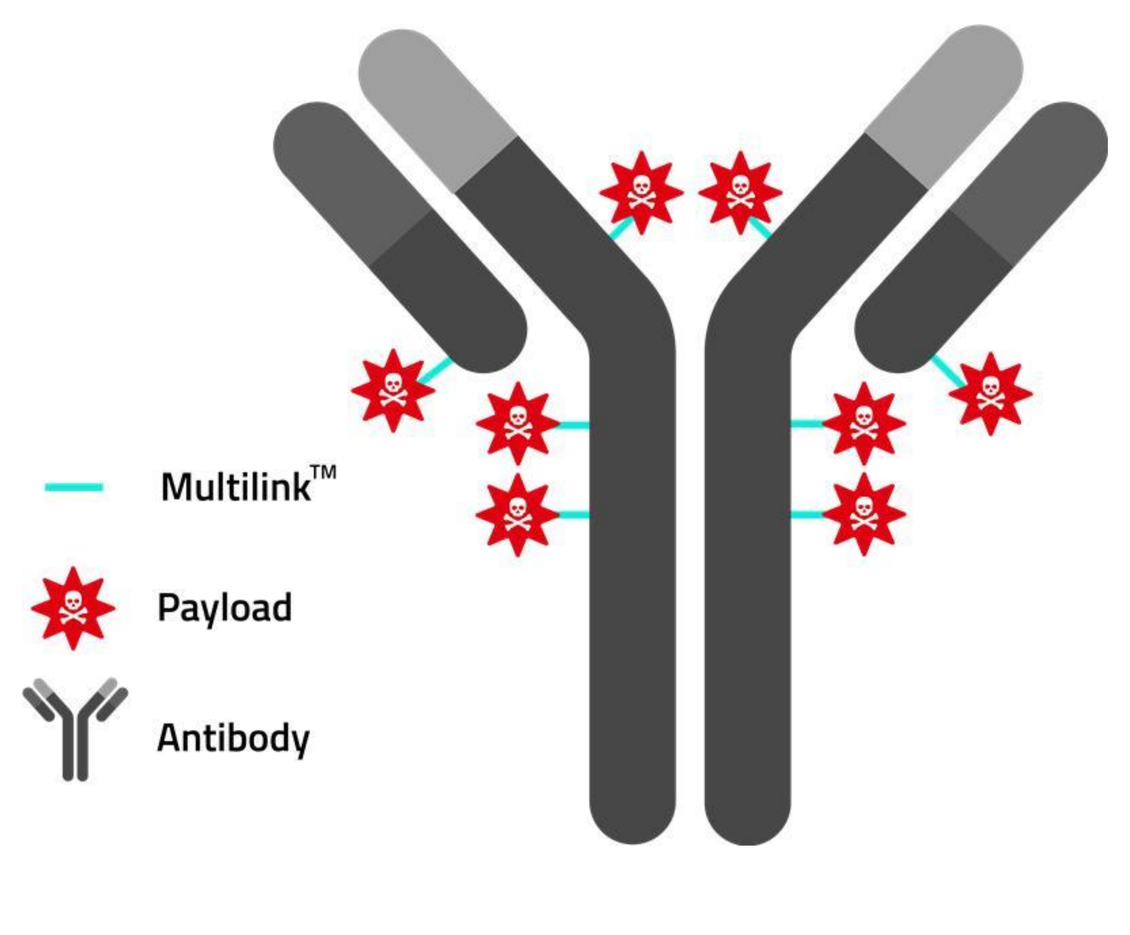
Multilink™
Payload
Antibody

METHODS

Mouse AML xenograft models. THP-1: NOD/SCID mice were inoculated with 1x10⁷ cells in tail vein. Mice were randomized based on body weight in groups of 10 mice, and treatment started seven day after cell inoculation. **MOLM-13 Luc:** NOD/SCID mice were inoculated with 2x10⁶ cells in tail vein. Mice were randomized based on total flux value 3 days after inoculation in groups of 8 mice and treatment started 7 days after inoculation. **MV4-11 Luc:** NCG mice were inoculated with 2x10⁷ cells in tail vein. Mice were randomized based on total flux value 14 days after inoculation in groups of 8 mice and treatment started the same day with azacitidine at 3.5mg/kg for 5 days, venetoclax at 100mg/kg for 14 days. Tumor growth was imaged twice per week after Luciferin administration. **OCI-AML-3 :** NOD-SCID mice were subcutaneously injected with 2x10⁷ cells. Debio 1562M was given once IV at 10mg/kg, azacitidine at 3.5mg/kg for 5 days, venetoclax at 100mg/kg for 14 days.
PDX mouse model: NOG or NOG-EXL mice were inoculated with 2x10⁶ AML cells from CTG-2239, CTG-2457, CTG-3930 and CTG-2227 in tail vein. Mice were randomized when at least 20% of human CD45 positive cells in the blood and bone marrow was reached. Treatment started the day of randomization and FACS analysis were performed 21 days after treatment start.
AML and MDS patient samples: blast cells were seeded in appropriate medium to follow blast viability in proliferative conditions. Staining of the cells with a cocktail of antibodies and annexin-V allowed to discriminate live and proliferative tumor cells with the PharmaFlow platform.
CD37 expression was determined by incubation of a dose range of naratuximab for 30 minutes at 4° C and detection with a AF488-labelled secondary Ab.
CD37 internalization was measured over 6 hours at 37° C with 5 µg/mL pHrodo dye coupled to Debio 1562M following manufacturer's instructions (Thermo Fisher P35355). For patient samples, 24h incubation with 10 µg/mL was used.
GLP toxicity study was performed in CD1 mice, with 2 iv administrations on days 1 and 21 of Debio 1562M at 50 mg/kg.
All in vivo studies were conducted in accordance with institutional guidelines and NCRI Guidelines for the welfare and use of animals in cancer research

ACKNOWLEDGEMENTS

To Alain Monjardet, René Wuttke, Marie-Claude Roubaudi-Fraschini, Camille Kowalski and Franck Brichory for their active contribution to this work.

RESULTS

CD37 is expressed on AML and MDS blasts from patient samples

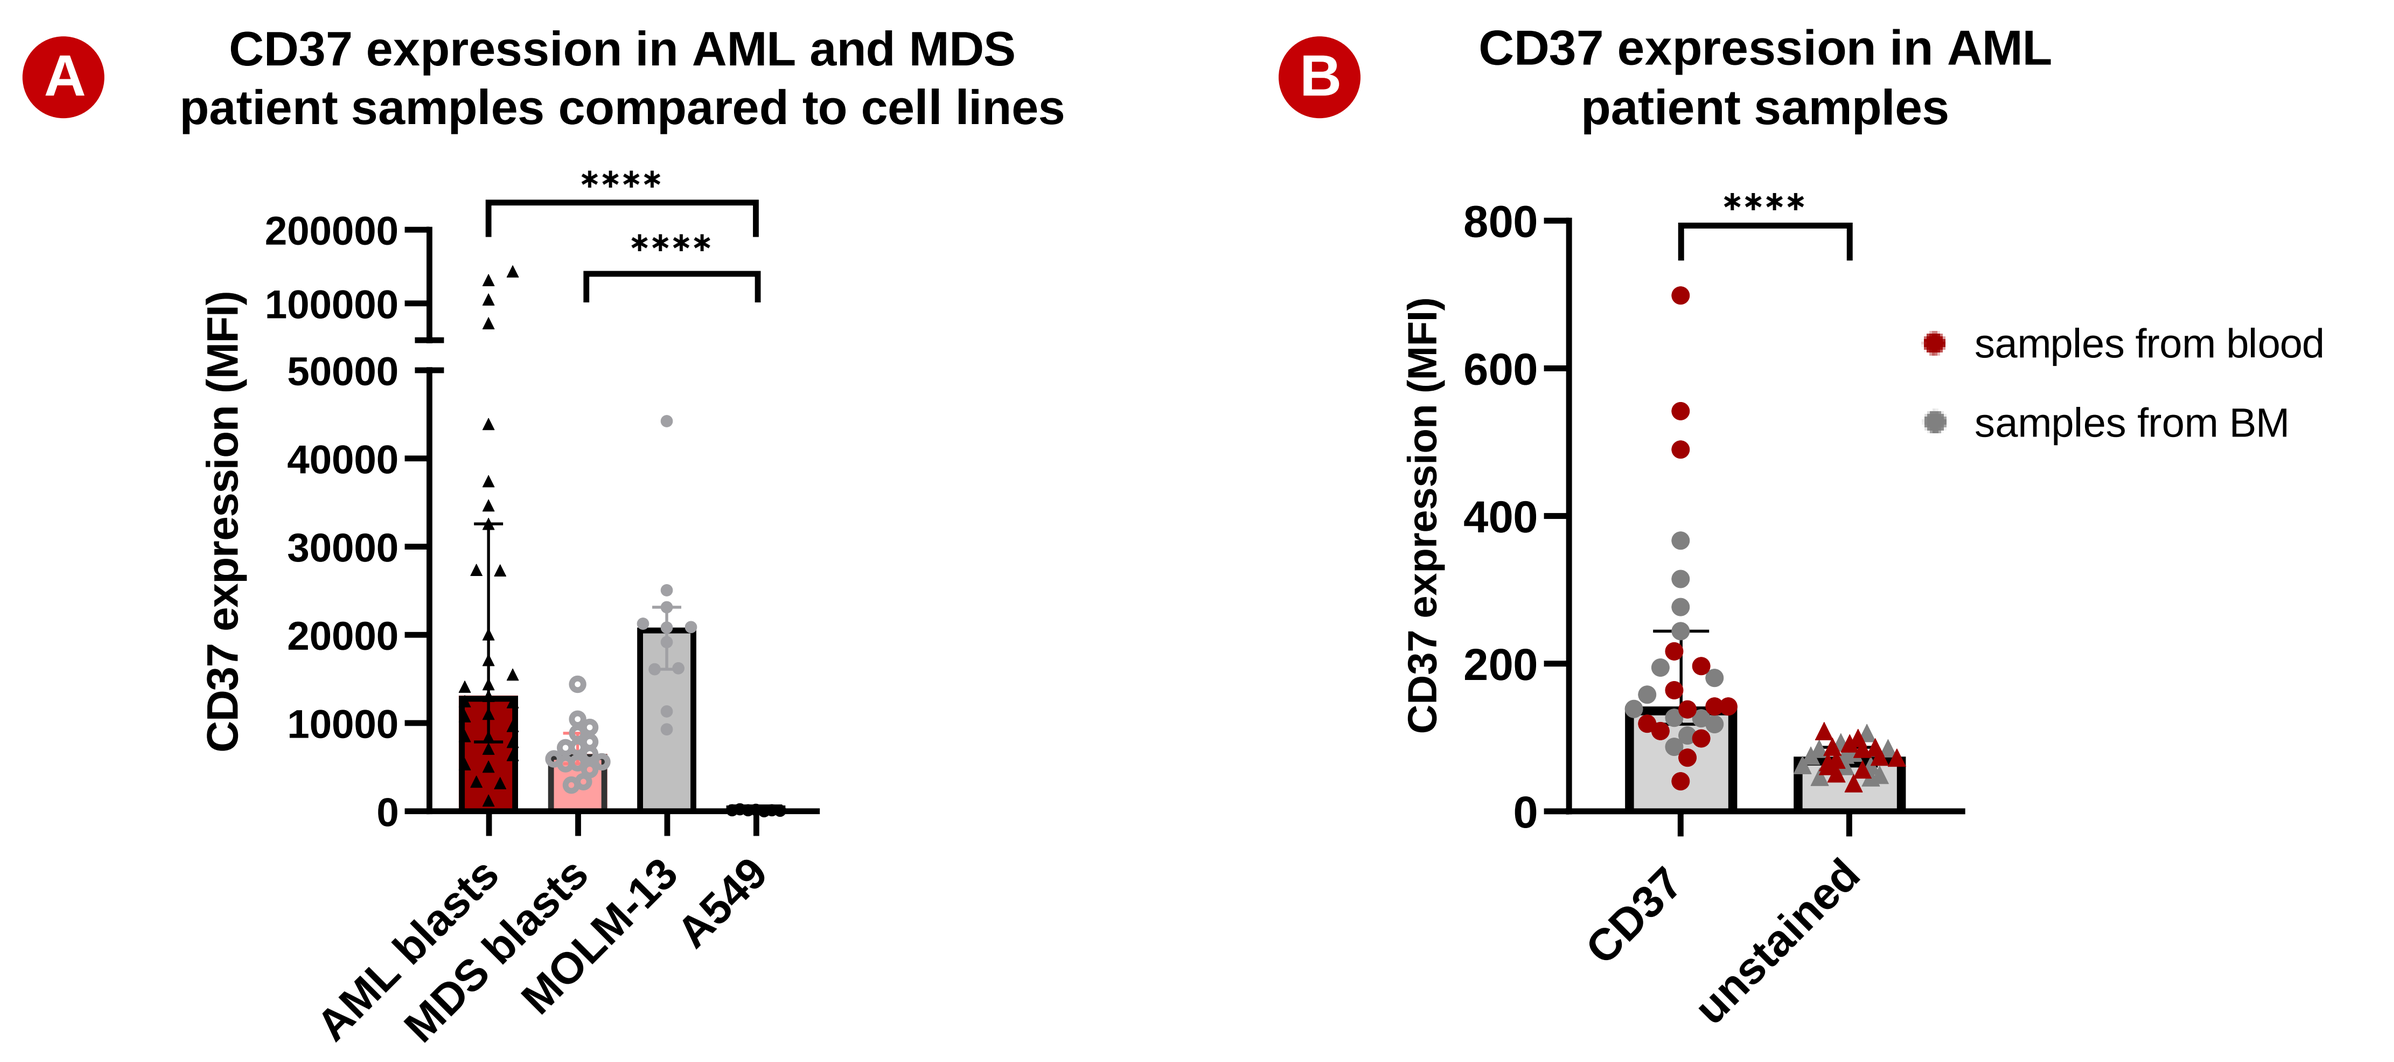
A
CD37 expression in AML and MDS patient samples compared to cell lines
CD37 expression (MFI)
AML blasts
MDS blasts
MOLM-13
A549
B
CD37 expression in AML patient samples
samples from blood
samples from BM
CD37
unstained

Figure 1. CD37 is detected on AML and MDS patient samples from different sources
A. CD37 mean fluorescence intensity (MFI) on primary bone marrow (BM) samples from 31 AML and 15 MDS patients compared to MOLM-13 AML cell line and A549 (CD37 negative) lung cancer cell line. B. CD37 MFI on primary samples from 13 BM or 14 whole blood of AML patients. Same samples with secondary only staining were used as negative controls (unstained).

Debio 1562M is better internalized in AML and MDS blasts compared to normal and malignant B cells and is highly cytotoxic

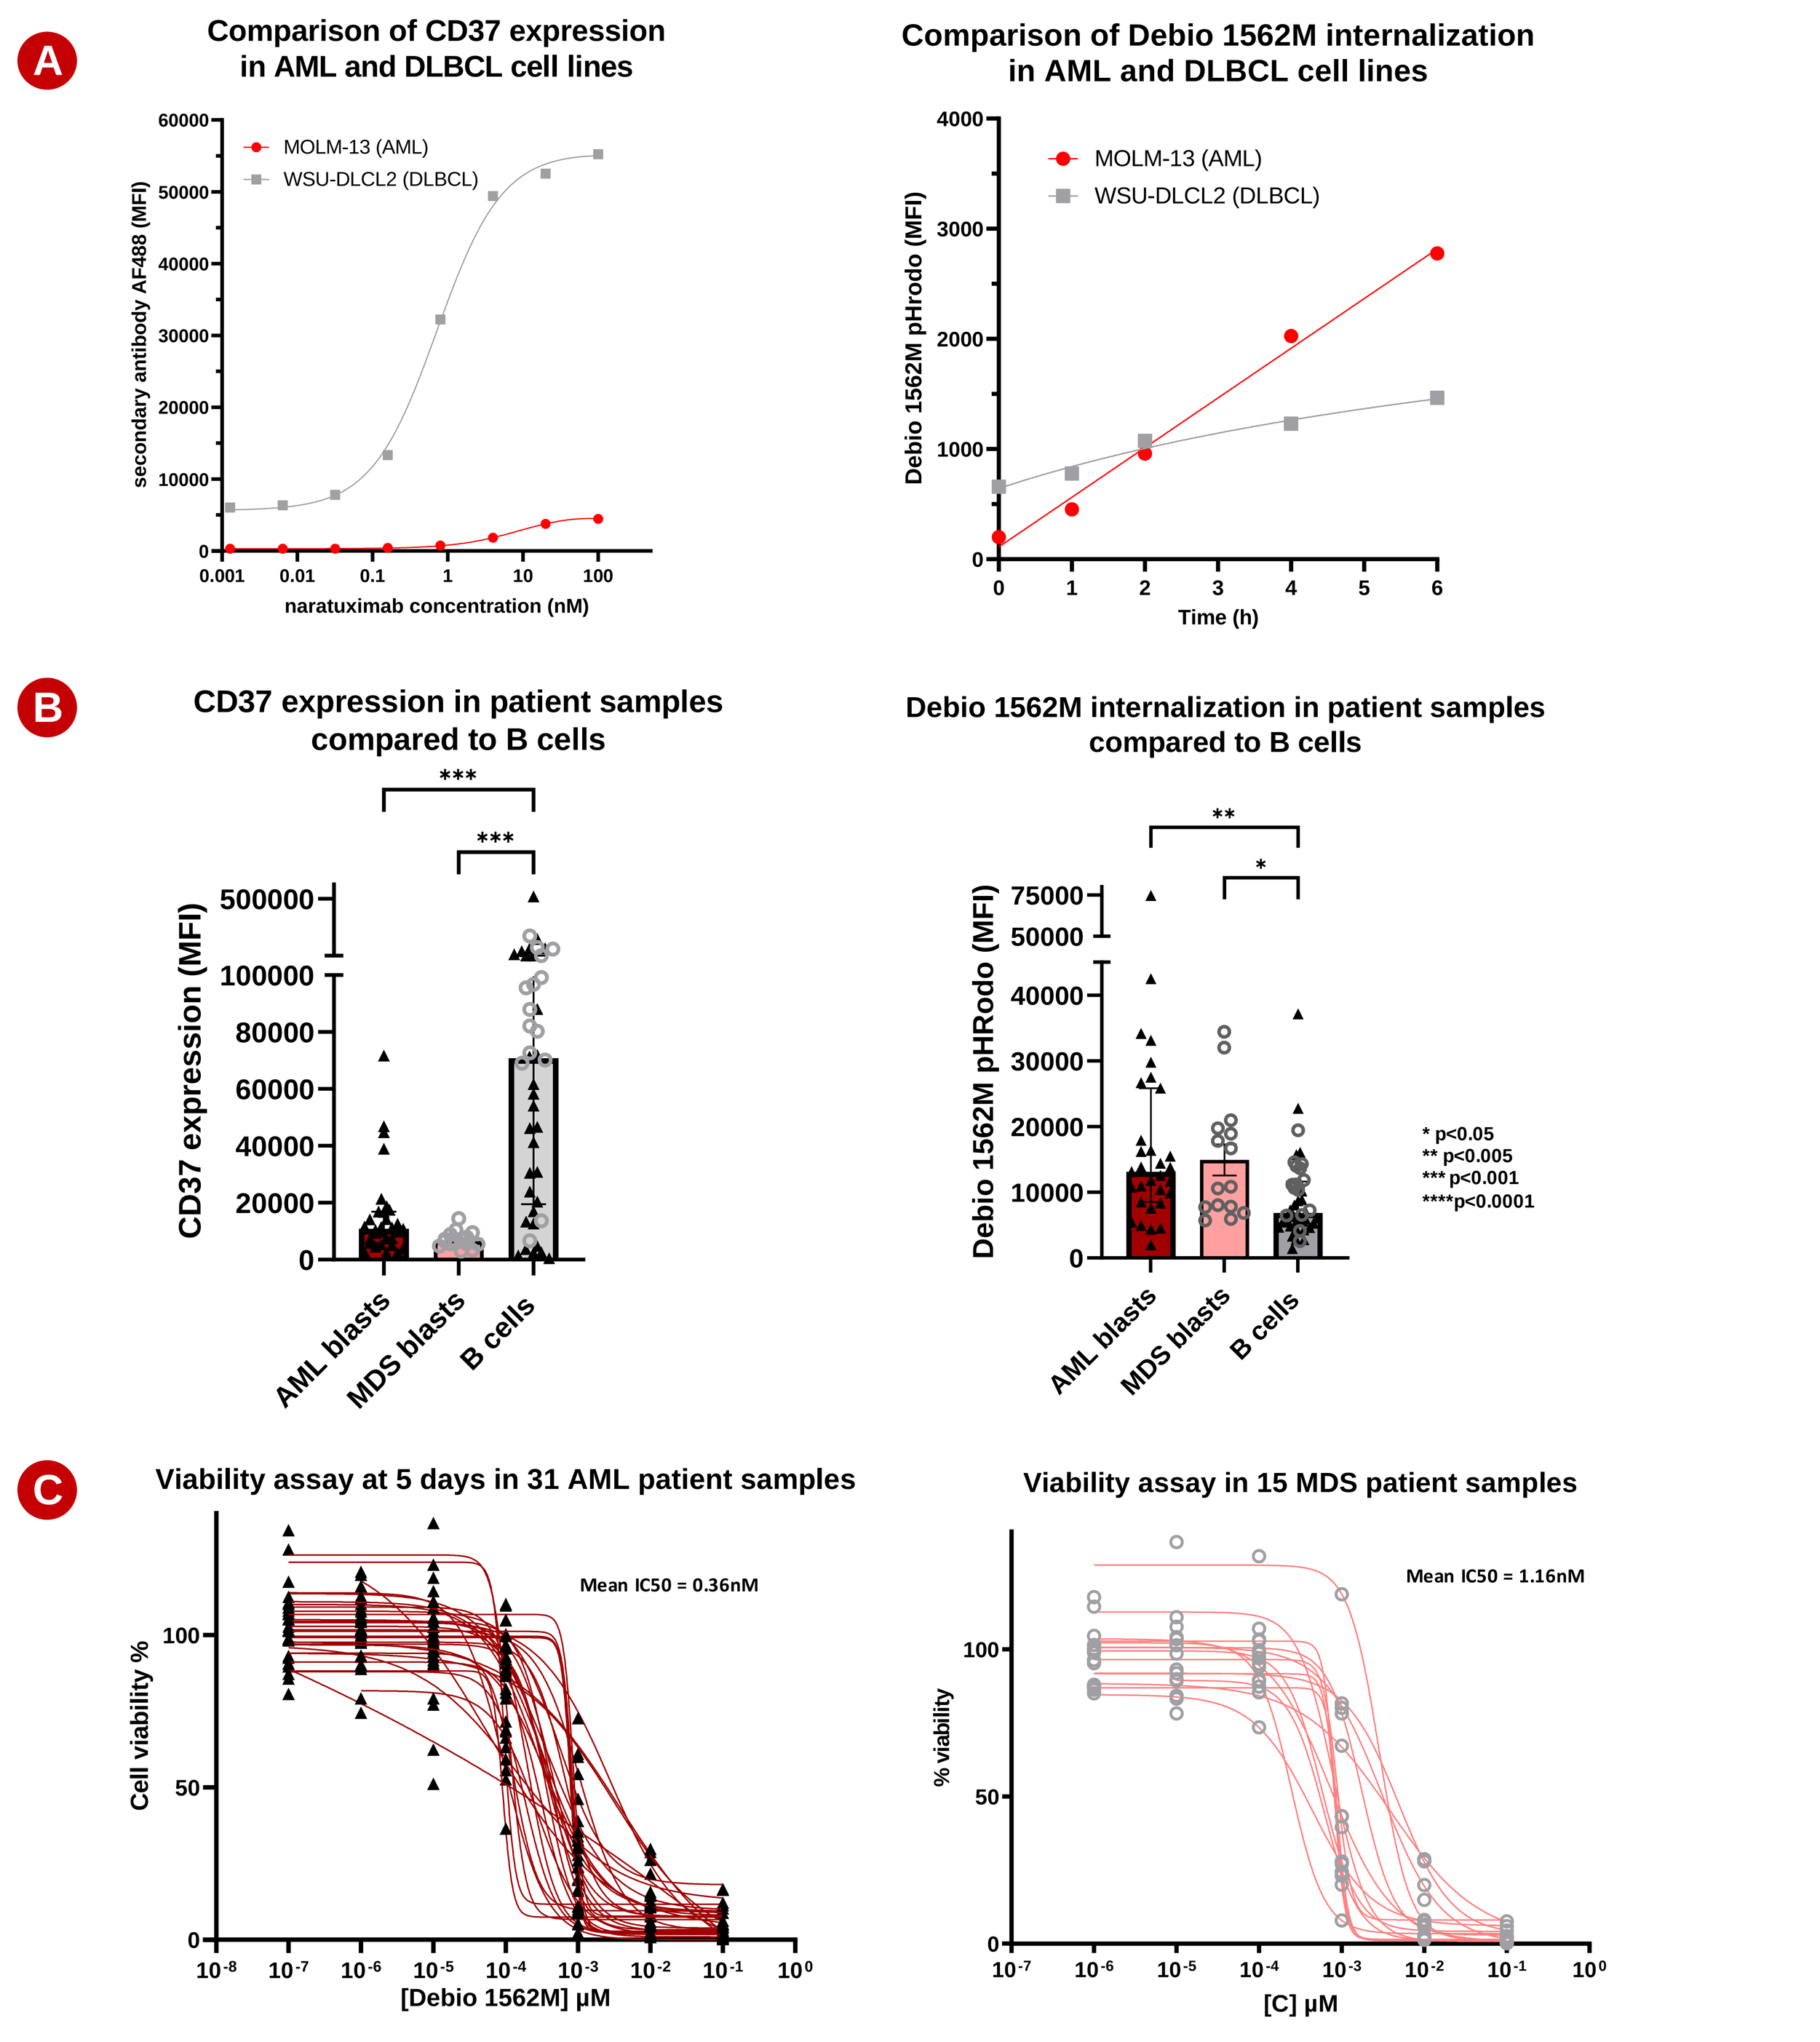
A
Comparison of CD37 expression in AML and DLBCL cell lines
MOLM-13 (AML)
WSU-DLCL2 (DLBCL)
naratuximab concentration (nM)
Comparison of Debio 1562M internalization in AML and DLBCL cell lines
Time (h)
B
CD37 expression in patient samples compared to B cells
Debio 1562M internalization in patient samples compared to B cells
AML blasts
MDS blasts
B cells
* p<0.05
** p<0.005
*** p<0.001
****p<0.0001
C
Viability assay at 5 days in 31 AML patient samples
Mean IC50 = 0.36nM
[Debio 1562M] µM
Viability assay in 15 MDS patient samples
Mean IC50 = 1.16nM
[C] µM

Figure 2. Low CD37 expression in AML/MDS versus B cells is compensated by efficient internalization and thus activity of Debio 1562M
A. WSU-DLCL2 DLBCL cell line has higher expression of CD37 than MOLM-13 AML cell line, however internalization of Debio 1562M is more efficient in MOLM-13 than in WSU-DLCL2 cell line. B. In AML and MDS blasts from patient samples, CD37 expression is lower than on healthy B cells coming from the same samples, however Debio 1562M internalization is more efficient in blasts. C. Efficient internalization is translated by strong cytotoxic activity of Debio 1562M in all AML and MDS blasts.

Debio 1562M significantly improves survival in AML CDX models

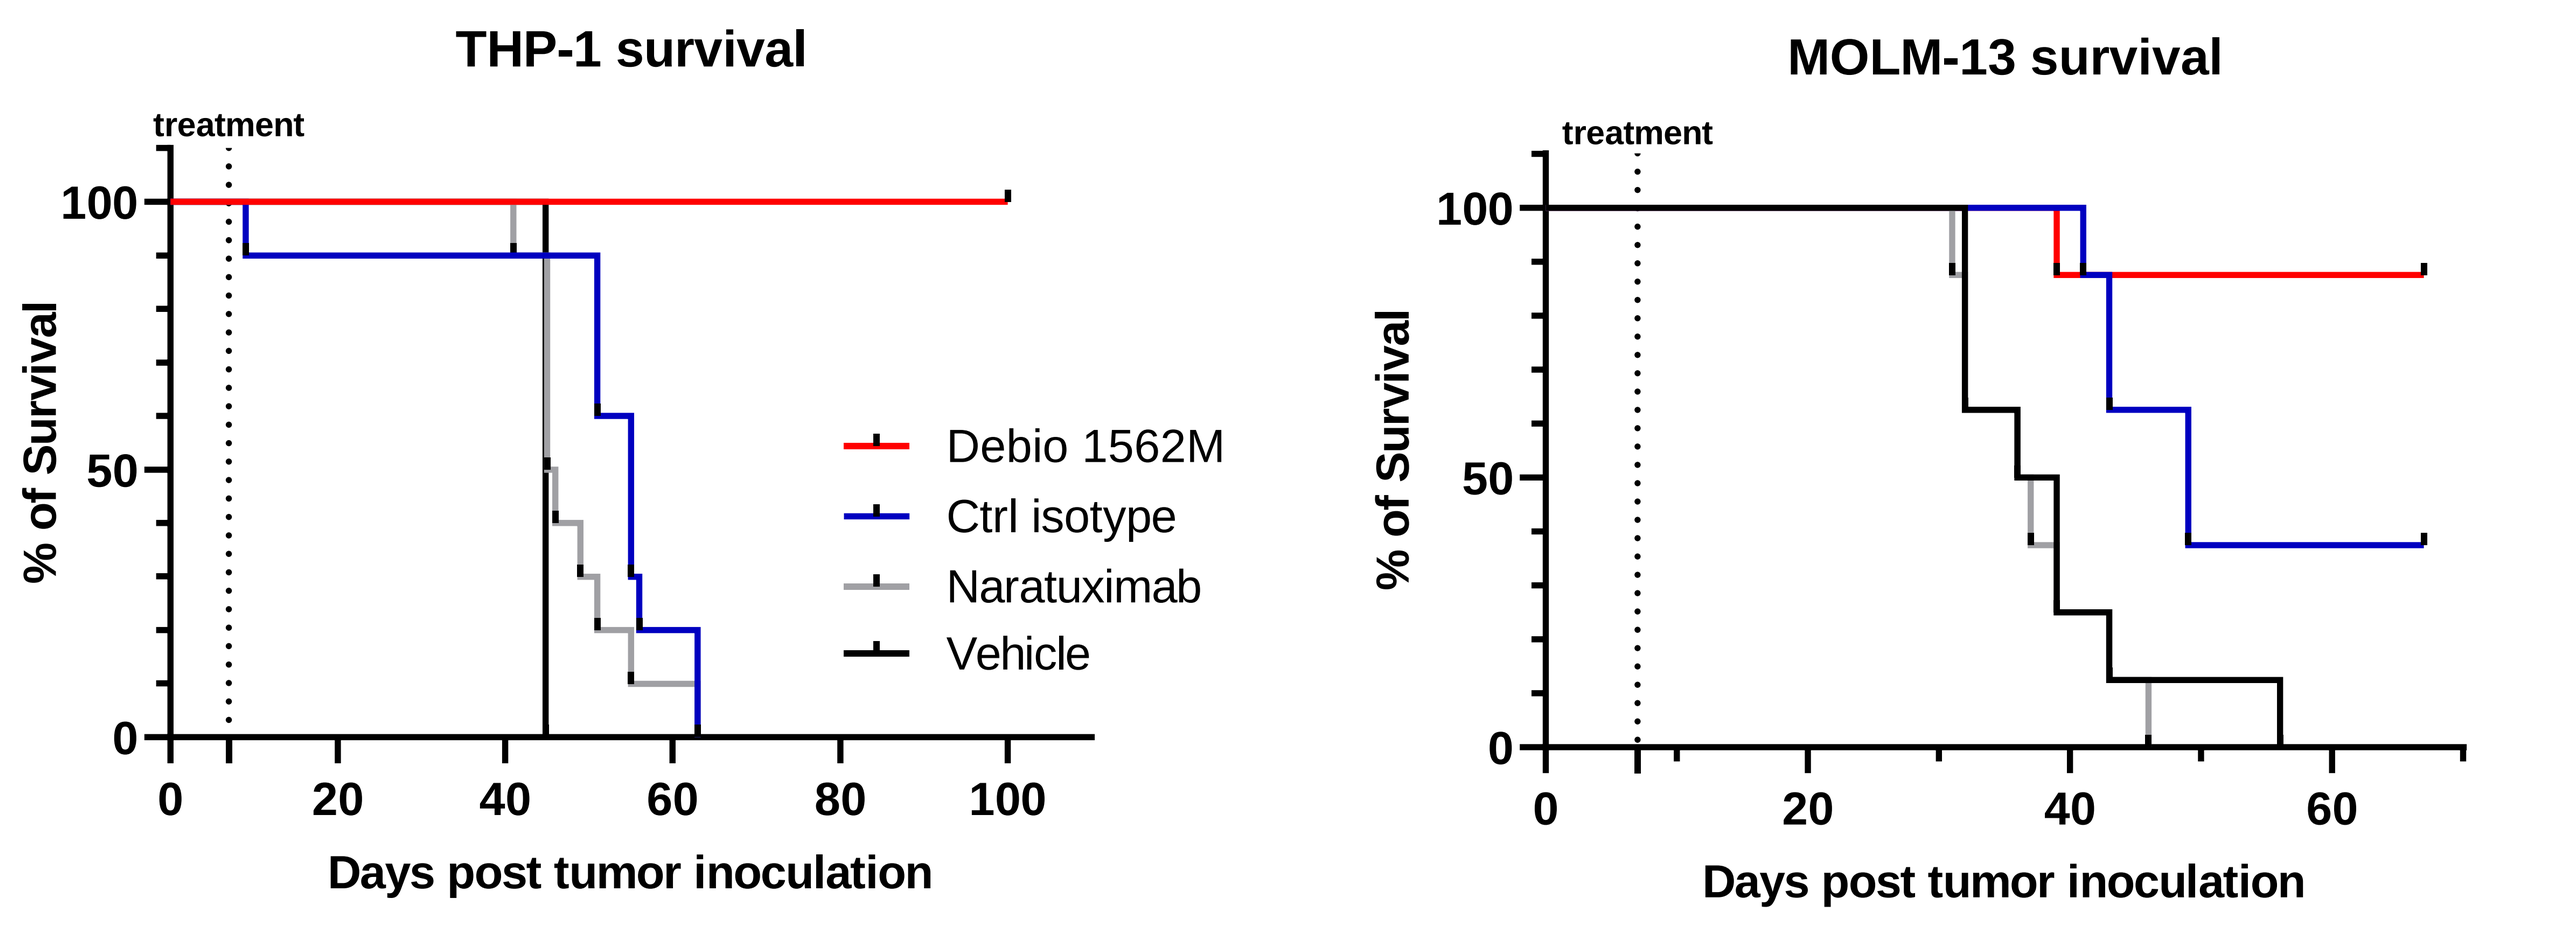
THP-1 survival
MOLM-13 survival
treatment
% of Survival
Debio 1562M
Ctrl isotype
Naratuximab
Vehicle
Days post tumor inoculation

Figure 3. in vivo efficacy of Debio 1562M in THP-1 and MOLM-13 disseminated models
Debio 1562M, control isotype (Trastuzumab-Multilink™-DM1) or naratuximab were administered once at 5 mg/kg. Specific and targeted activity of Debio 1562M is demonstrated in both models compared to naratuximab or control isotype.

Debio 1562M anti-tumoral activity is superior to venetoclax+azacitidine standard of care in preclinical models

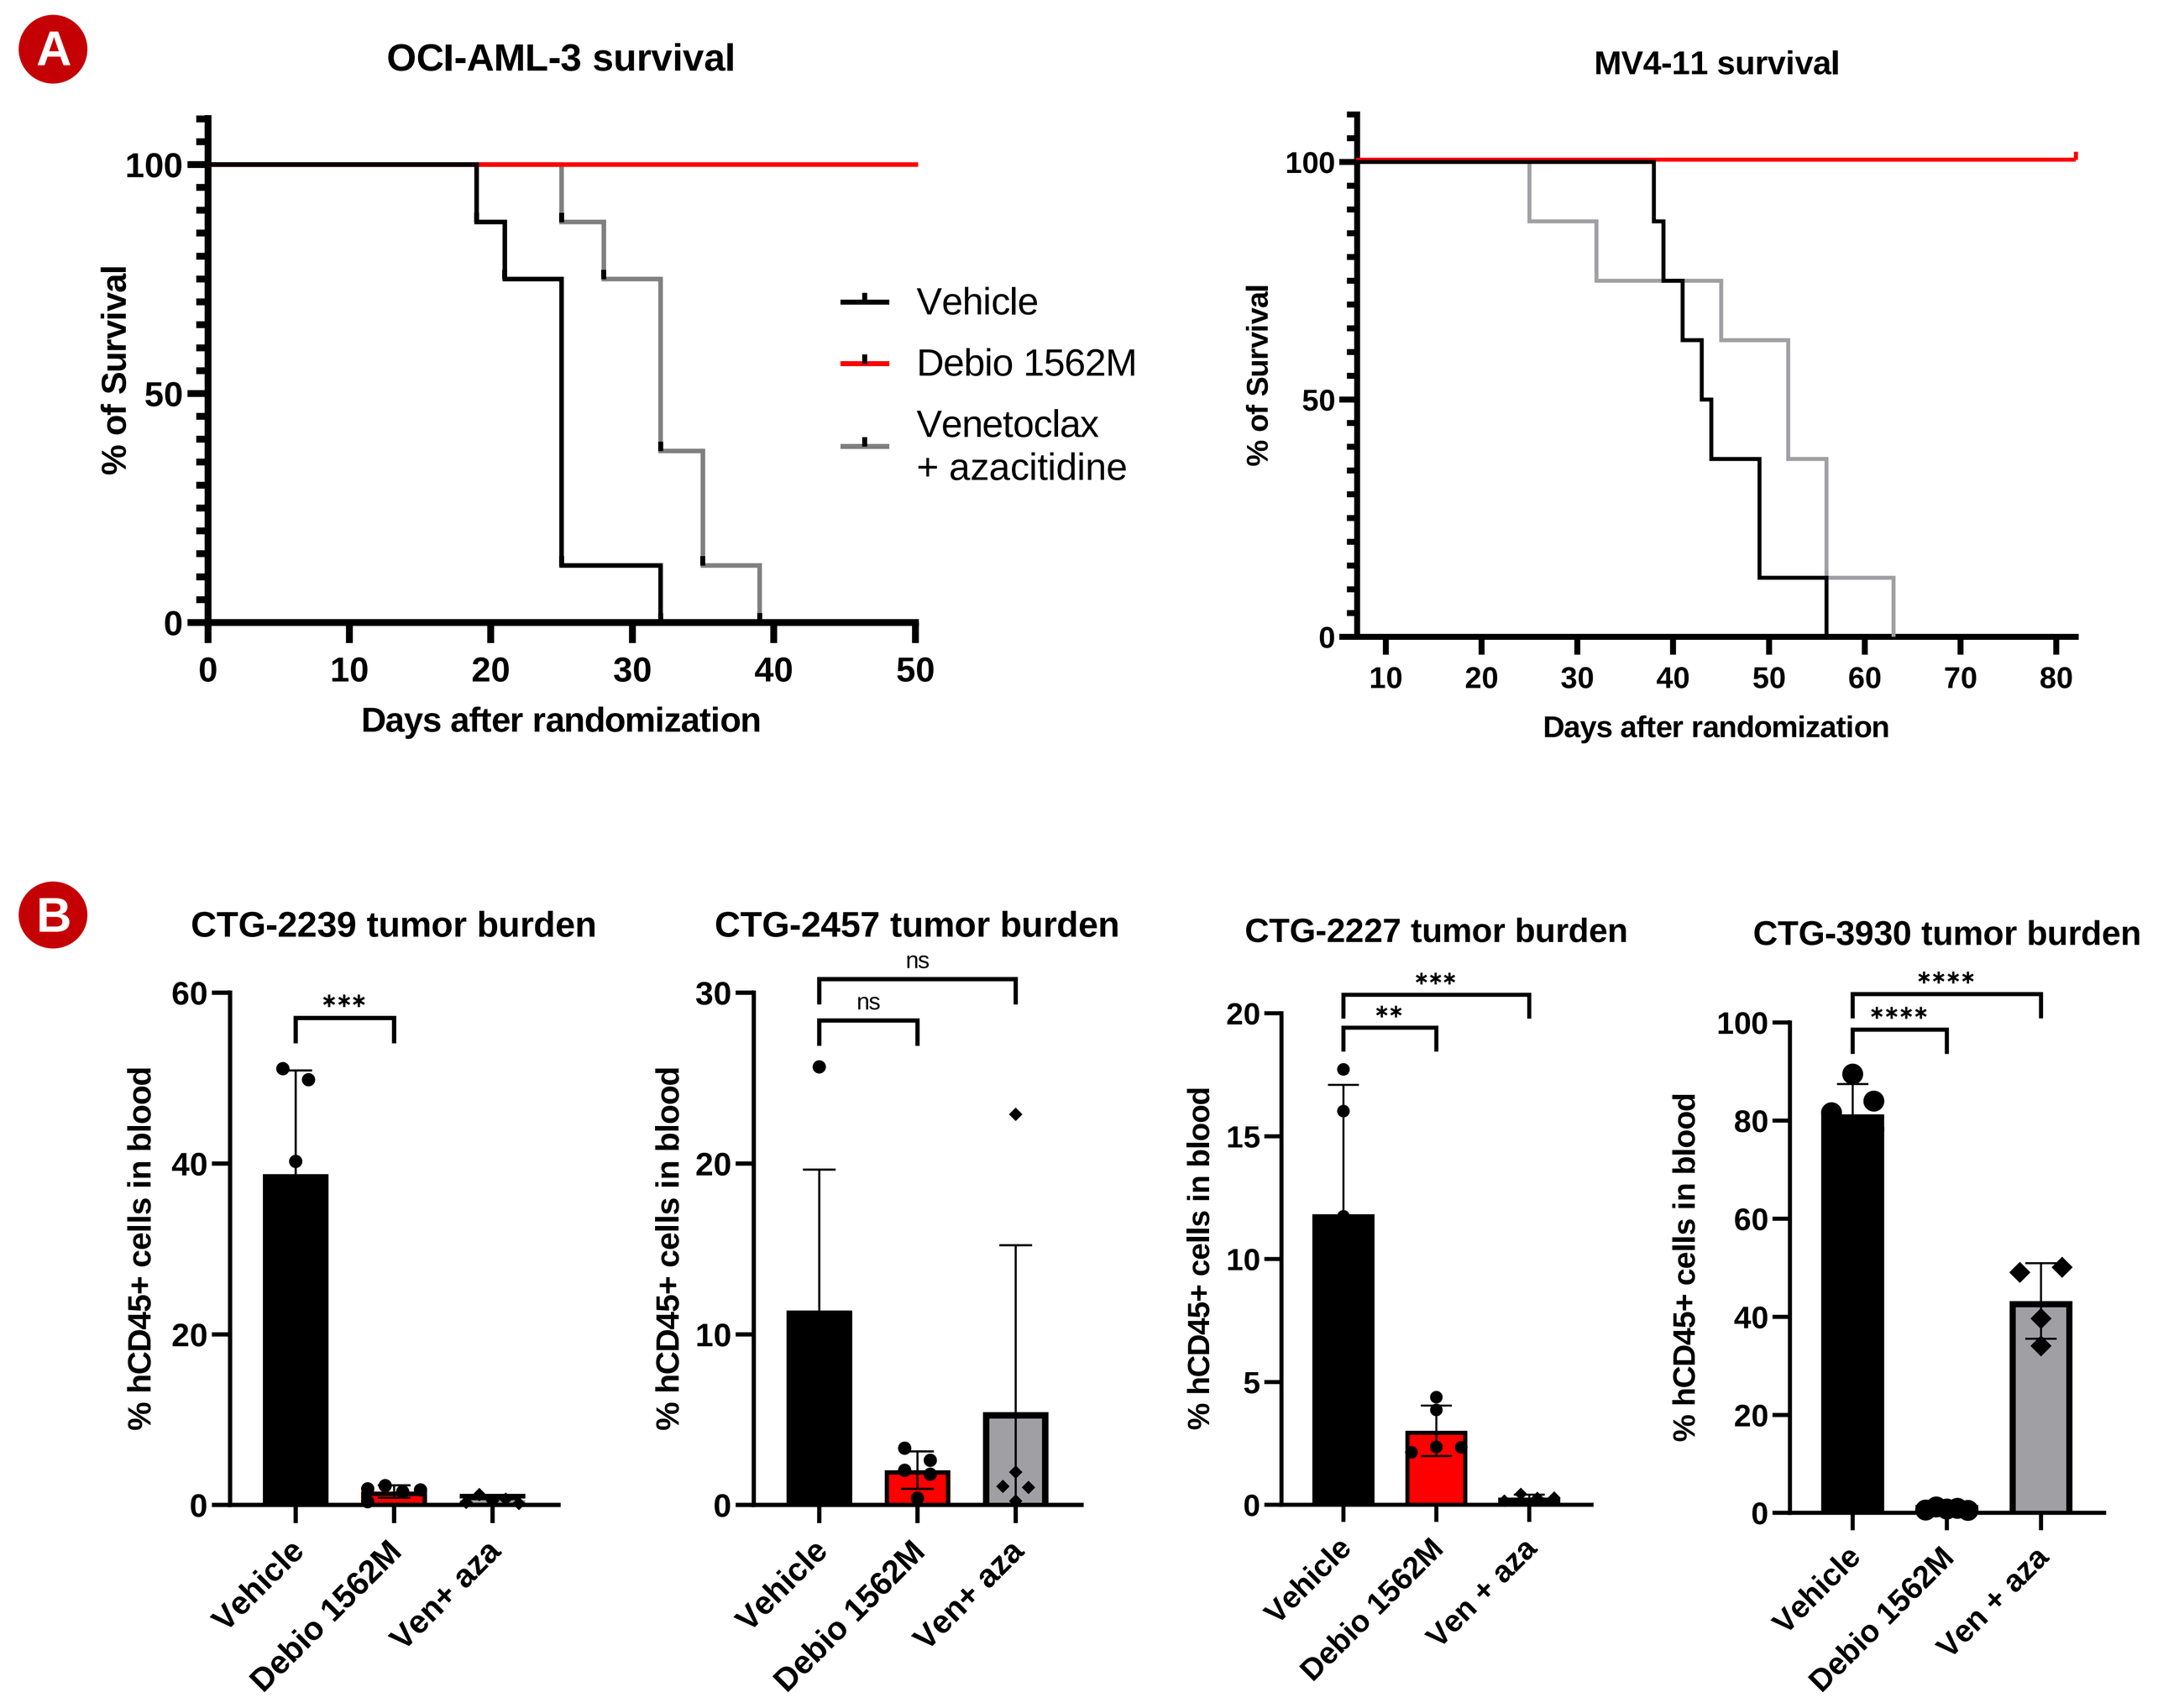
A
OCI-AML-3 survival
MV4-11 survival
Vehicle
Debio 1562M
Venetoclax + azacitidine
Days after randomization
B
CTG-2239 tumor burden
CTG-2457 tumor burden
CTG-2227 tumor burden
CTG-3930 tumor burden
% hCD45+ cells in blood
Vehicle
Debio 1562M
Ven+ aza

Figure 4. in vivo efficacy of Debio 1562M compared to venetoclax + azacitidine in CDX and PDX models
A. Debio 1562M administered once at 5 mg/kg (MV4-11) or 10mg/kg (OCI-AML-3) significantly improves survival compared to venetoclax + azacitidine (venetoclax QDx15, 100mg/kg and azacitidine QDx5, 3.5mg/kg). B. In PDX models, Debio 1562M administered once at 10 mg/kg is equivalent to venetoclax+ azacitidine (venetoclax QDx21, 100mg/kg and azacitidine 5on2off x3, 2.5mg/kg) to reduce tumor blasts number (measured 21 days after treatment start). Debio 1562M is also very active in CTG-3930 venetoclax+ azacitidine resistant model.

Debio 1562M has a significant safety margin in mice with a toxicology profile related to payload's known toxicities

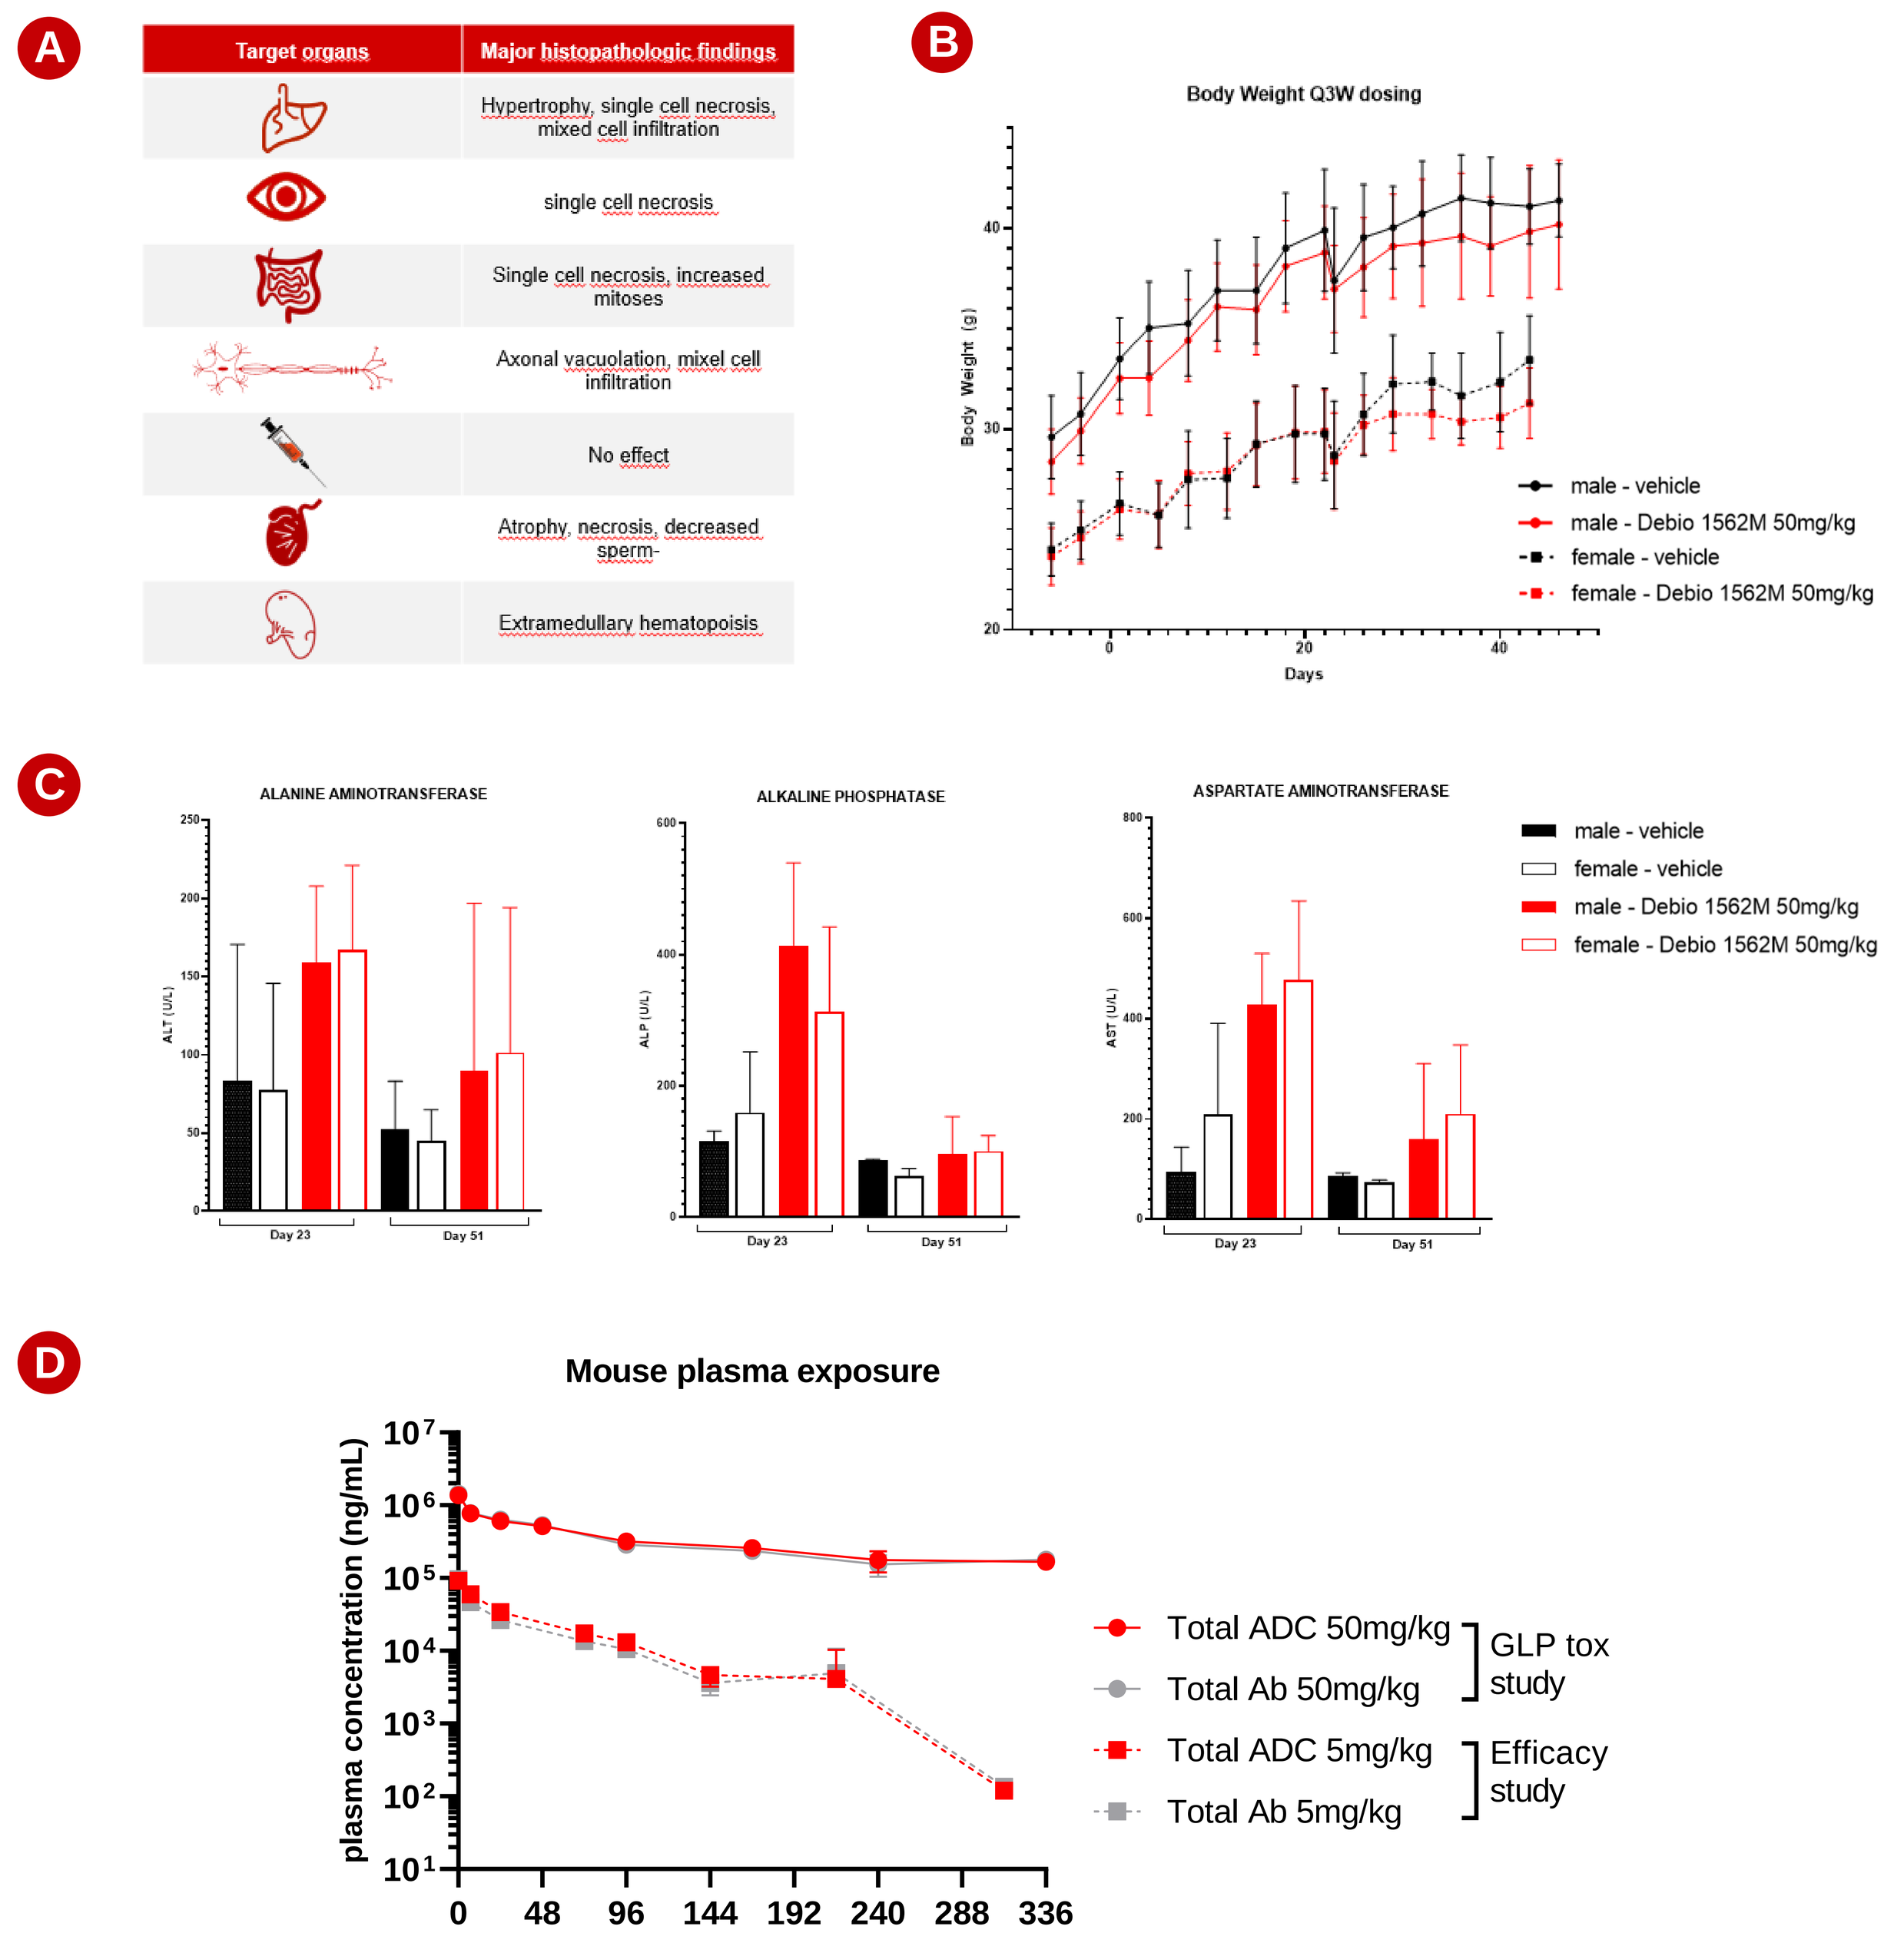
A
Target organs
Major histopathologic findings
Hypertrophy, single cell necrosis, mixed cell infiltration
single cell necrosis
Single cell necrosis, increased mitoses
Axonal vacuolation, mixel cell infiltration
No effect
Atrophy, necrosis, decreased sperm-
Extramedullary hematopoisis
B
Body Weight Q3W dosing
male - vehicle
male - Debio 1562M 50mg/kg
female - vehicle
female - Debio 1562M 50mg/kg
C
ALANINE AMINOTRANSFERASE
ALKALINE PHOSPHATASE
ASPARTATE AMINOTRANSFERASE
D
Mouse plasma exposure
plasma concentration (ng/mL)
Time (h)
Total ADC 50mg/kg
Total Ab 50mg/kg
GLP tox study
Total ADC 5mg/kg
Total Ab 5mg/kg
Efficacy study

Study / Dose				Safety Margins in mice allowing tumor regression	
GLP Tox / 50 mg/kg Q3W		Efficacy MOLM-13 / 5 mg/kg Q3W			
AUClast (µg.h/mL)	Cmax (µg/mL)	AUClast (µg.h/mL)	Cmax (µg/mL)	**AUClast**	**Cmax**
92,900	1,310	4,030	92	**23**	**14**

Figure 5. Debio 1562M is tolerated at 50mg/kg in GLP toxicity study in mice
Debio 1562M is not cross reactive with mouse CD37, therefore on-target toxicity is not evaluated. A. Summary of off-target organ toxicities after 2 administrations (Q3W). B. Debio 1562M has no impact on body weight in males or females compared to control group. C. Liver enzymes were slightly elevated at the end of the treatment phase and were in line with control data at the end of the recovery. D. Comparison of exposure at 50mg/kg in the GLP toxicity study and exposure at 5mg/kg in MOLM-13 tumor bearing mice allows to calculate a safety margin between 14 and 23. PK profile of total ADC and total Ab are overlayed, highlighting the excellent stability of the ADC in plasma.

REFERENCES

(1)Frigault et al, Blood 2021
(2)Steinsland et al., Blood 2020
(3)Brobowicz et al, Int. J. of Molecular Sciences 2020
(4)Beckwith et al, Leukemia. 2014 Jul;28(7):1501-10.
(5)Deckert et al, Blood. 2013 Nov 14;122(20):3500-10
(6)Pereira et al, Mol Cancer Ther. 2015 Jul;14(7):1650-60
(7)Yan et al, iScience. 2021 Oct 9;24(11):103249
(8)Zhang et al, Biosci Rep. 2020 May 29;40(5):BSR20200008;
(9)Larkin et al, ASH 2018 poster 616, Blood vol 132, Sup 1, 29 November 2018,

CONCLUSIONS

- **CD37 is broadly expressed in AML and MDS**
- **Debio 1562M ADC targeting CD37 is efficiently internalized in AML and MDS primary tumor cells and highly cytotoxic**
- **Debio 1562M monotherapy improves survival in several AML CDX models compared to SoC and significantly reduces tumor burden in PDX models**
- **Overall Debio 1562M activity and safety in preclinical models is promising for future clinical development in AML and MDS**

CONTACT

Debiopharm International S.A.
Lausanne, Switzerland
www.debiopharm.com
Lisa.ivanschitz@debiopharm.com